RAMP 205: A phase 1b/2a study of gemcitabine, nab-paclitaxel, avutometinib, and defactinib in untreated metastatic pancreatic ductal adenocarcinoma

Presented at
AACR Pancreatic Cancer 2023, Boston, MA
ABSTRACT C003

Kian-Huat Lim[1], Manuel Hidalgo[2], Mark H. O'Hara[3], Kristen R. Spencer[4], Ignacio Garrido-Laguna[5], David G. DeNardo[1], Vijeta Bhambhani[6], Gloria Patrick[6], Yaofeng Cheng[6], Silvia Coma[6], Jonathan A. Pachter[6], Louis J. Denis[6]

[1]Division of Oncology, Department of Medicine, Washington University in St. Louis, St. Louis, MO; [2]Division of Hematology and Medical Oncology, Weill Cornell Medical College, New York City, NY; [3]Division of Hematology-Oncology, Department of Medicine, Abramson Cancer Center, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA; [4]Department of Medicine, Perlmutter Cancer Center of NYU Langone Health, New York, NY; [5]Division of Medical Oncology, Huntsman Cancer Institute, Salt Lake City, UT; [6]Verastem Oncology, Needham, MA.

Background

Pancreatic Ductal Adenocarcinoma (PDAC)

- PDAC accounts for ~90% of pancreatic cancer.[1-3]
- Majority of PDAC (52%) is initially diagnosed with advanced/inoperable disease.[1-3]
- mOS for untreated Stage IV PDAC is 4.5 mo, with 5-year survival for metastatic disease <3%.[1-3]
- Activating *KRAS* mutations (mt) occur in almost all of PDAC.[4]

RAMP 205 Study Rationale

- **Avutometinib**: First-in-class oral RAF/MEK clamp that potently inhibits MEK kinase activity while also blocking the compensatory reactivation of MEK by upstream RAF (**Figure 1A**).[5-8]
- **Defactinib**: Selective inhibitor of focal adhesion kinase (FAK), a target which has been shown to mediate resistance to multiple anticancer agents (**Figure 1A**).[9-12]
- Preclinically, FAKi reduces stromal density in PDAC.[13]
- Whereas avutometinib + FAKi induced tumor inhibition and increased survival, addition of chemotherapy induced tumor regression and further improved survival in a *KRAS/TP53* mt PDAC mouse model (**Figure 1B**).[14-15]
- Clinically, defactinib + gemcitabine + pembrolizumab was safe, showed responses, decreased stromal density, and increased CD8+ T cell infiltration in PDAC (NCT02546531).[16]

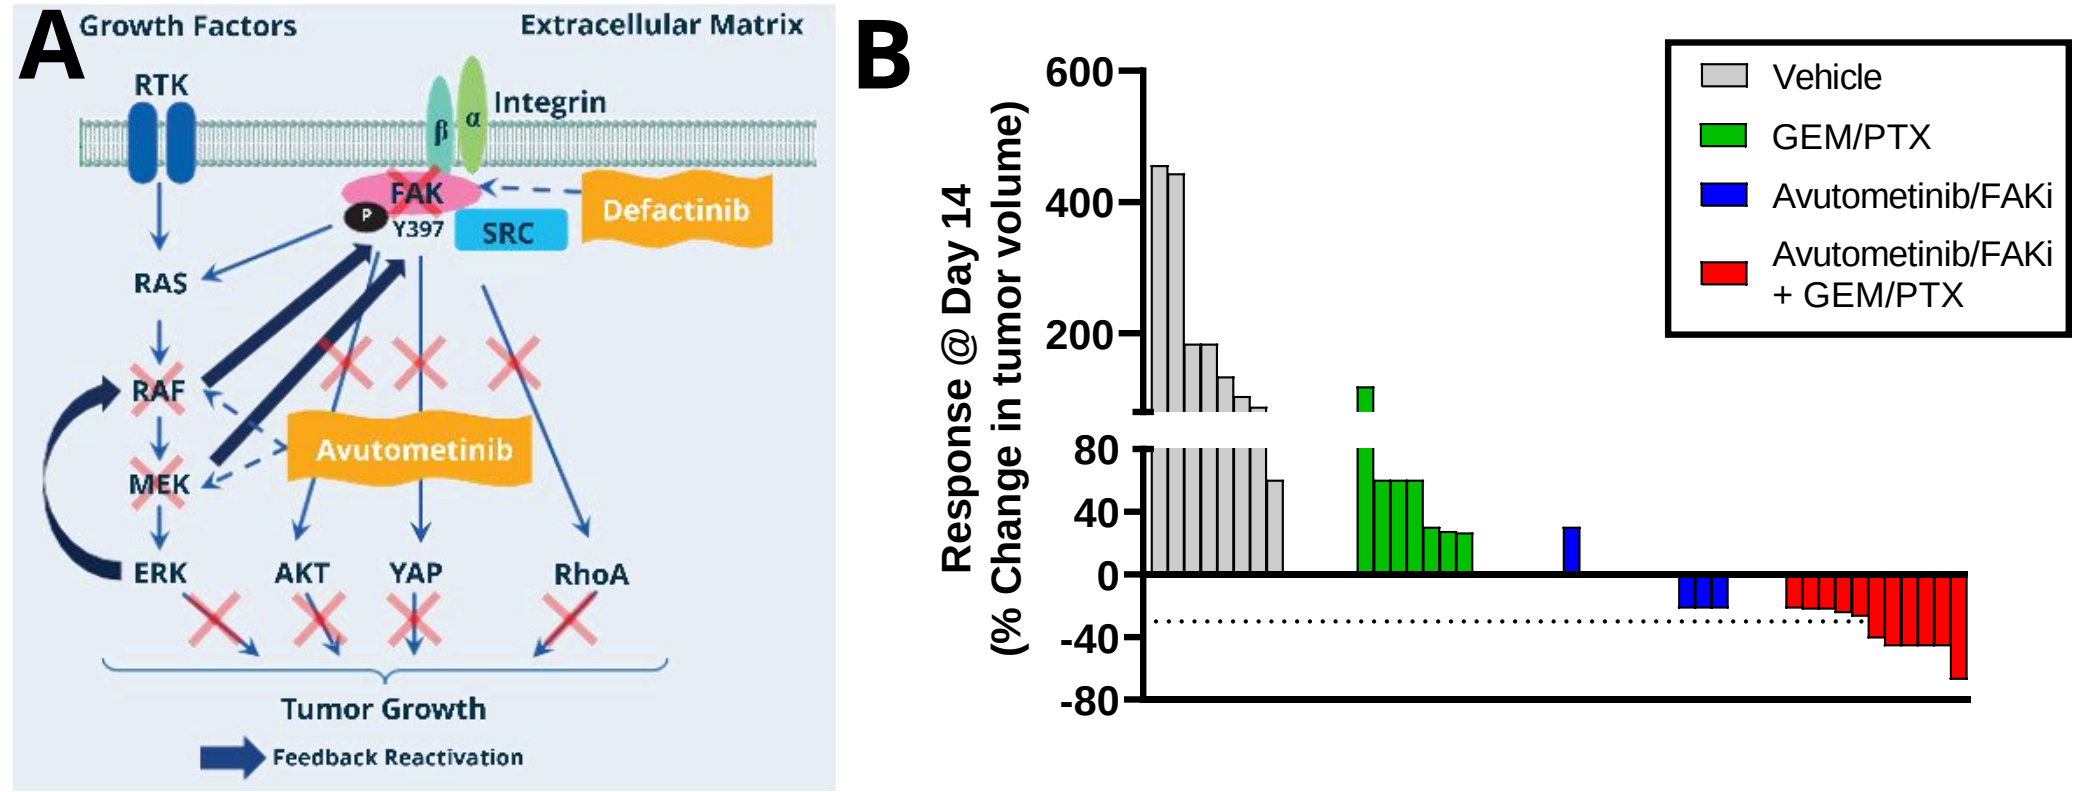

A
Growth Factors
Extracellular Matrix
RTK
Integrin
FAK
Y397
SRC
Defactinib
RAS
RAF
MEK
Avutometinib
ERK
AKT
YAP
RhoA
Tumor Growth
Feedback Reactivation
B
Response @ Day 14 (% Change in tumor volume)
600
400
200
80
40
0
-40
-80
Vehicle
GEM/PTX
Avutometinib/FAKi
Avutometinib/FAKi + GEM/PTX

Figure 1 (A) Scientific rationale for avutometinib and FAKi combination. (B) Changes in tumor volume in KP2 KRAS G12D/p53 pancreatic cancer tumor-bearing mice treated with avutometinib (0.3 mg/kg PO, QD 5 days on/2 days off) + FAKi (50 mg/kg PO, BID 5 days on 2 days off) ± gemcitabine/paclitaxel (GEM/PTX; 75 mg/kg/5 mg/kg IV every 5 days; x5) are shown. Paclitaxel was used as a surrogate for nab-paclitaxel in mice. N = 10 mice/group. *ERK, extracellular signal-regulated kinase; FAK, focal adhesion kinase; FAKi, FAK inhibitor; MEK, mitogen-activated protein kinase; RAF, rapidly accelerated fibrosarcoma; RTK, receptor tyrosine kinase; YAP, yes-associated protein.*

RAMP 205 Study Summary

RAMP 205 Study Design

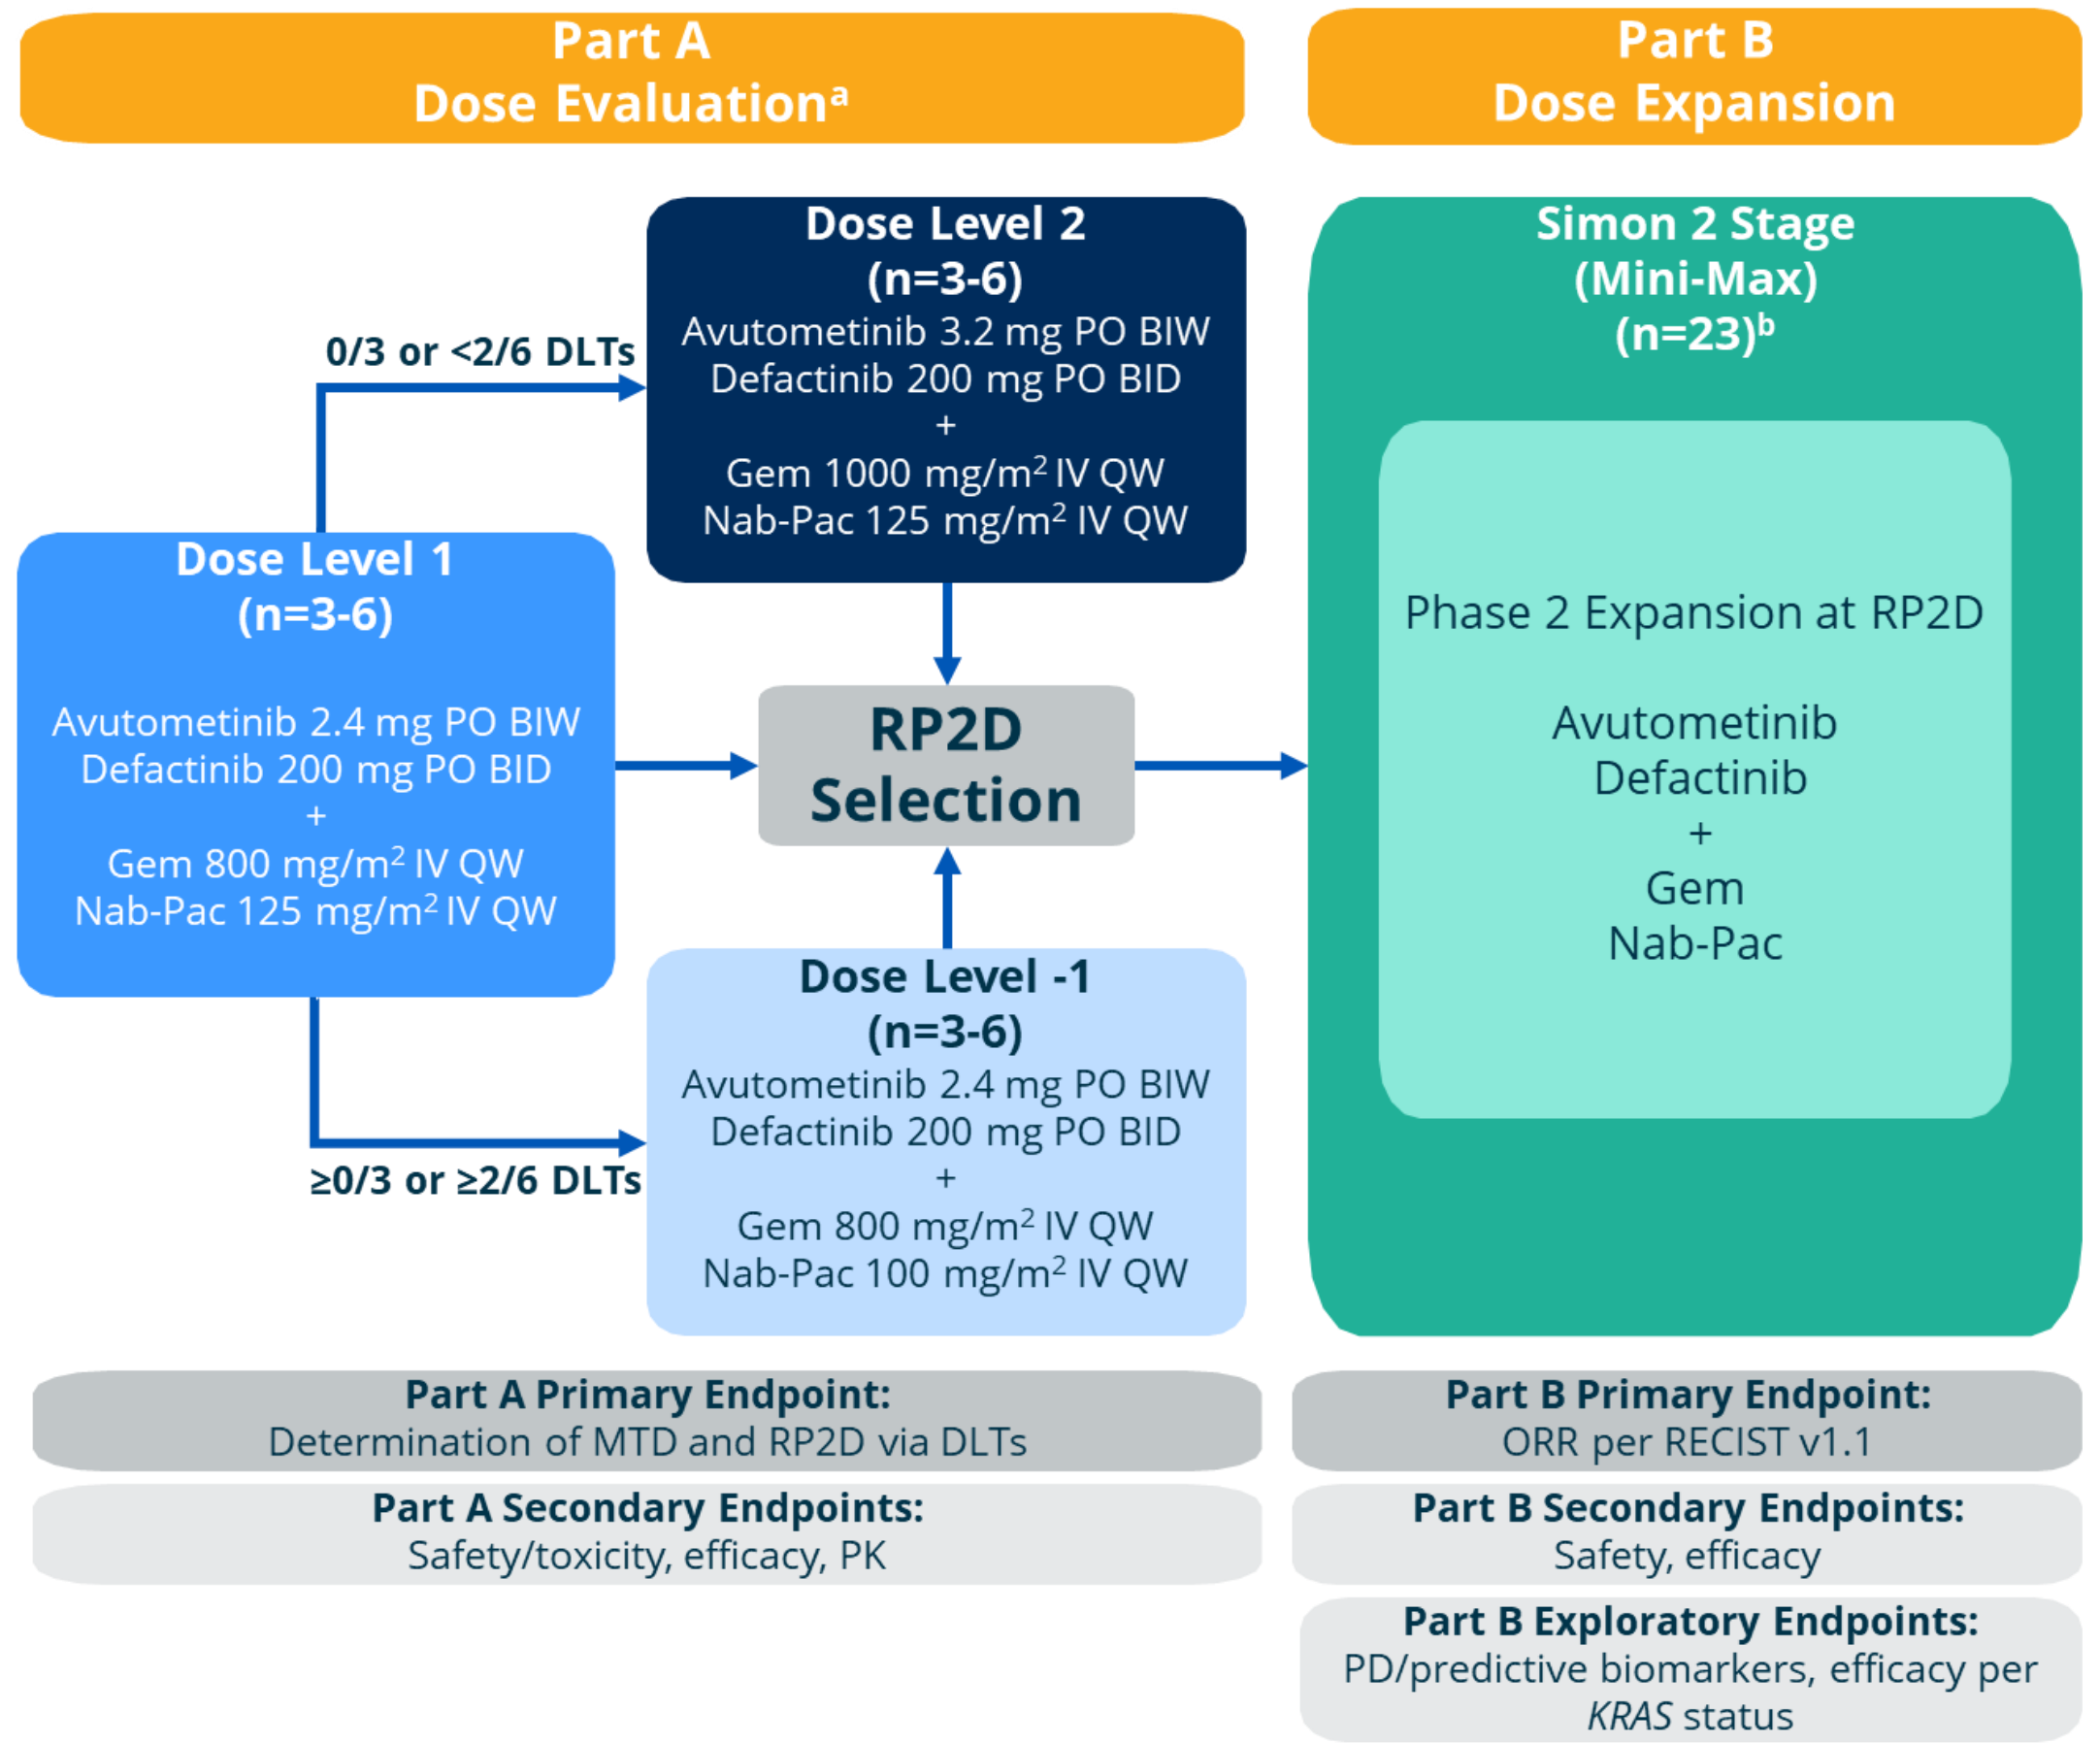

Part A
Dose Evaluation[a]
Part B
Dose Expansion
Dose Level 2
(n=3-6)
Avutometinib 3.2 mg PO BIW
Defactinib 200 mg PO BID
+
Gem 1000 mg/m² IV QW
Nab-Pac 125 mg/m² IV QW
0/3 or <2/6 DLTs
Dose Level 1
(n=3-6)
Avutometinib 2.4 mg PO BIW
Defactinib 200 mg PO BID
+
Gem 800 mg/m² IV QW
Nab-Pac 125 mg/m² IV QW
RP2D Selection
Dose Level -1
(n=3-6)
Avutometinib 2.4 mg PO BIW
Defactinib 200 mg PO BID
+
Gem 800 mg/m² IV QW
Nab-Pac 100 mg/m² IV QW
≥0/3 or ≥2/6 DLTs
Simon 2 Stage
(Mini-Max)
(n=23)[b]
Phase 2 Expansion at RP2D
Avutometinib
Defactinib
+
Gem
Nab-Pac
Part A Primary Endpoint:
Determination of MTD and RP2D via DLTs
Part A Secondary Endpoints:
Safety/toxicity, efficacy, PK
Part B Primary Endpoint:
ORR per RECIST v1.1
Part B Secondary Endpoints:
Safety, efficacy
Part B Exploratory Endpoints:
PD/predictive biomarkers, efficacy per KRAS status

Figure 2 RAMP 205 study design. All study treatment administered 21/28 days. [a]Additional intermittent dose levels and alternate dosing schedules may be considered per emerging clinical data. [b]Approximately 23 evaluable patients will be enrolled in Part B (29 patients required for the Simon 2-stage minimax approach less the evaluable patients treated at the RP2D in Part A [n=6]).*BID, twice daily; BIW, twice weekly; DLTs, dose limiting toxicities; Gem, gemcitabine; IV, intravenous; MTD, maximum tolerated dose; Nab-Pac, nab-paclitaxel; ORR, objective response rate; PD, pharmacodynamics; PK, pharmacokinetics; PO, orally; pts, patients; QW, once weekly; RECIST v1.1, response evaluation criteria in solid tumors version 1.1; RP2D, recommended phase 2 dose.*

- **RAMP 205** is a multicenter, open-label, single arm Phase 1b/2a study of gemcitabine and nab-paclitaxel in combination with avutometinib and defactinib in patients with previously untreated metastatic PDAC (NCT05669482) (**Figure 2**).
- RAMP 205 will evaluate addition of avutometinib + defactinib to standard gemcitabine/nab-paclitaxel with the objective of increasing ORR and survival (N = ~35):
 - **Part A:** Dose evaluation (n = 6-12)
 - **Part B:** Dose expansion at RP2D (n = 23).
- Mandatory tumor biopsies and blood samples will be collected for correlative studies, including circulating tumor DNA, molecular profiling and PD markers.

Key Inclusion Criteria

1. Male or female patients ≥ 18 years of age.
2. Histologic/cytologic confirmed metastatic PDAC.
3. Measurable disease per RECIST v1.1
4. ECOG performance status of 0-1.
5. Eligible for treatment in 1L setting with standard gemcitabine and nab-paclitaxel (i.e. no prior systemic therapy for advanced/metastatic disease). Prior adjuvant/neoadjuvant therapy permitted if last intervention/dose ≥ 12 mo prior to metastatic disease diagnosis.
6. Willingness to provide tissue and blood samples (mandatory for part B only). Optional research biopsy in Part A may have been collected at the time of diagnostic biopsy.
7. Adequate material for central laboratory confirmation of *KRAS* mt status (does not need to be completed prior to enrollment).

Key Exclusion Criteria

1. Pancreatic neuroendocrine tumors, islet cell tumors, mucinous cystic, squamous or adenosquamous histologies.
2. Prior or concomitant treatment for metastatic PDAC. Palliative irradiation of lesions permitted as long as it is not considered a target lesion.
3. Prior treatment with RAS/MAPK or FAK inhibitors.
4. Presence of symptomatic ascites.
5. Major surgery within 4 weeks or minor surgery within 2 weeks of first dose of study intervention.
6. Leptomeningeal or symptomatic brain metastases unless patient has completed and recovered from treatment
7. Active skin disorder that has required systemic therapy within the past 1 year.
8. History of rhabdomyolysis.
9. Concurrent ocular disorders.

RAMP 205 Status

- As of September 2023, enrollment of RAMP 205 Part A in metastatic PDAC is ongoing (dose evaluation).
- Currently, there are 7 activated and 5 planned sites (12 total) in the United States (**Figure 3**)
- Please contact Verastem Oncology (**clinicaltrials@verastem.com**) for more details and refer to clinicaltrials.gov for up-to-date study information.

- We thank the patients, their families, and the trial teams at participating centers.
- Supported by Pancreatic Cancer Action Network (Therapeutic Accelerator Award).

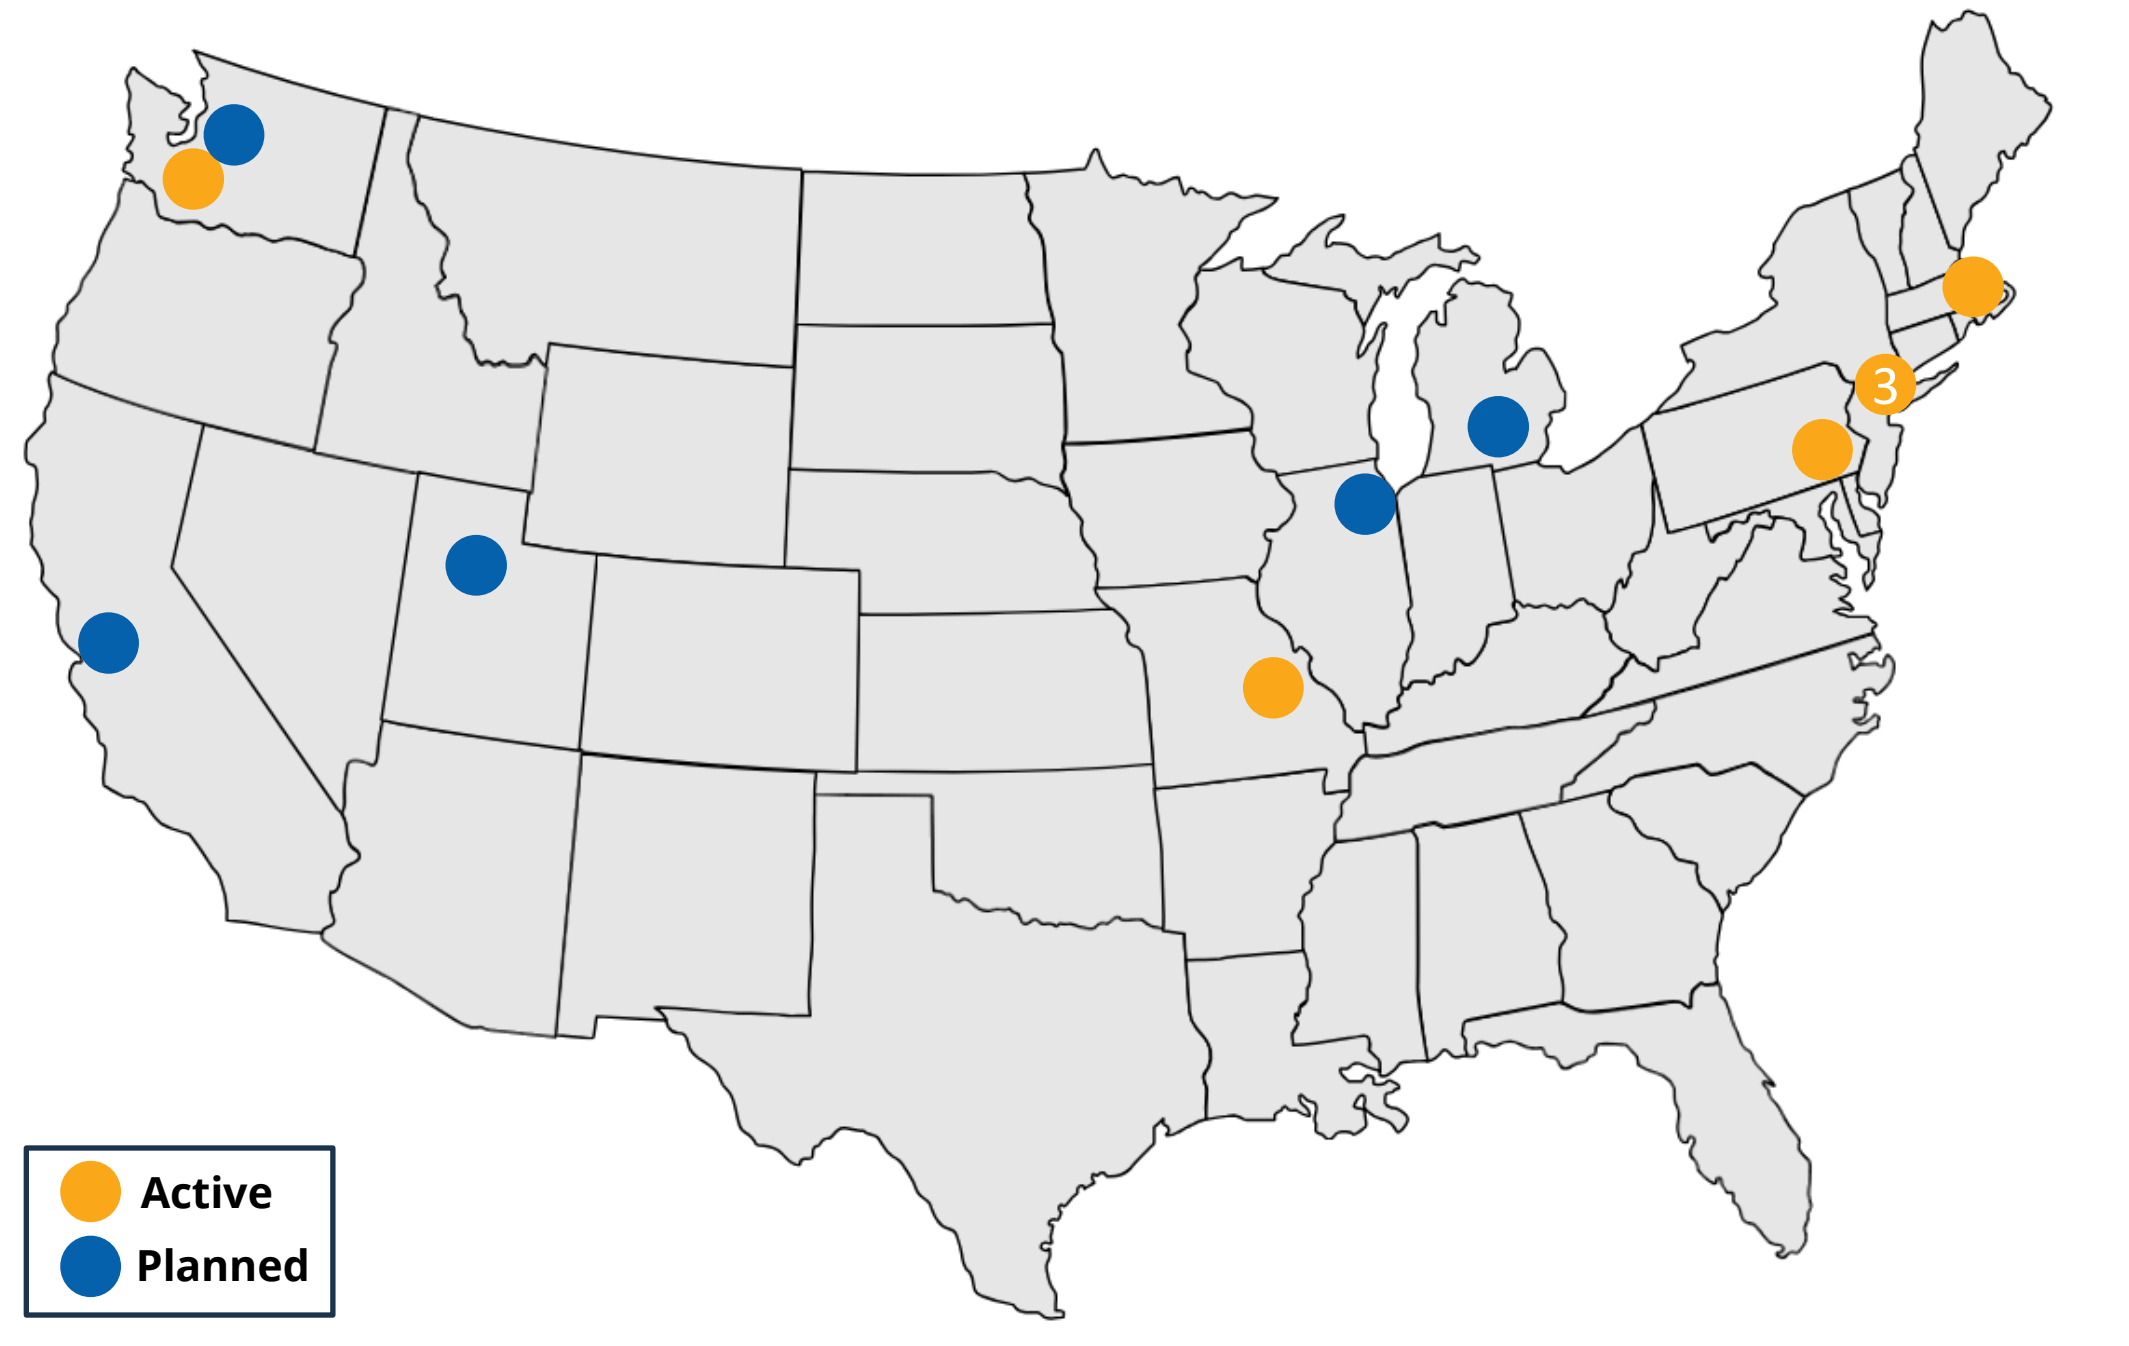

3
Active
Planned

Figure 3 Current RAMP 205 active and planned sites in United States.

References

1. American Cancer Society. *Facts & Figures 2023*.
2. National Cancer Institute Surveillance, Epidemiology, and End Results Program. Cancer Stat Facts: Pancreatic Cancer 2023.
3. Werner et al. *Nat Rev Clin Oncol* 2013;10:323-333.
4. Cox AD et al. *Nat Rev Drug Discov* 2014;13(11):828-851.
5. Martinez-Garcia C, et al. Clin Cancer Res. 2012;18:4806-4819.
6. Ishii N, et al. *Cancer Res*. 2013;73:4050-4060.
7. Lito P, et al. Cancer Cell. 2014;25:697-710.
8. Gonzalez-Del Pino GL, et al. *PNAS*. 2021;118:e2107207118.
9. Dawson JC, et al. *Nat Rev Cancer*. 2021;21:313-324.
10. Shinde R, et al. *Cancer Res*. 2020;80(Suppl 16):CT143.
11. Chen G, et al. *Mol Cancer Ther*. 2018;17:806-813.
12. Kang Y, et al. *J Natl Cancer Inst*. 2013;105(19):1485-95.
13. Jiang et al. *Nat Med*. 2016;22:851-860.
14. Coma S et al. The 4th RAS Initiative Symposium; 17-19Oct2022.
15. Coma S et al. AACR Special Conference: Pancreatic Cancer; 25-27Sep 2023.
16. Wang-Gillam et al. *Clin Cancer Res*. 2022;28(24):5254-5262.