FAK/PYK2 Inhibitors Sensitize Hypoxia-Induced Drug Resistant Multiple Myeloma Cells

Barbara Muz[1], Maurizio Buggio[1], Feda Azab[1], Anthony F. Trombino[2], Mahesh Padval[2] and Abdel Kareem Azab[1]

[1]Department of Radiation Oncology, Cancer Biology Division, Washington University in Saint Louis School of Medicine, Saint Louis MO, USA; [2] Verastem, Inc., Boston MA, USA

Introduction

Multiple myeloma (MM) is a plasma cell malignancy, characterized by plasma cell accumulation in the bone marrow (BM) and hyperproduction of immunoglobulin G (IgG). Despite the implementation of novel therapies, more than 70% of MM patients relapse due to drug resistance and minimal-residual disease (MRD) attributed to cancer stem cells. The hypoxic nature of the BM plays a critical role in MM cells acquiring a stem cell-like phenotype, and together with cellular and acellular components of the BM microenvironment contributes to drug resistance leading to MRD. In this study, we tested inhibition of proline-rich tyrosine kinase 2 (PYK2), using the dual FAK (focal adhesion kinase)/PYK2 inhibitors VS-4718 and VS-6063, on reversing the hypoxia-inducible stem cell-like phenotype of MM cells and sensitizing them to therapy both *in vitro* and *in vivo*.

Methods

MM cell lines (MM1s, RPMI8226 and H929) were treated with the FAK/PYK2 inhibitors VS-4718 or VS-6063 obtained from Verastem, Inc., in the presence or absence of bortezomib under normoxic (21% O_2) and hypoxic (1% O_2) conditions. MM cells were analyzed for cell proliferation/survival by using an MTT assay; apoptosis was analyzed by Annexin V/PI staining and analyzed by flow cytometry, and cell signaling associated with cell apoptosis was analyzed by western blotting. Furthermore, we tested the effect of the PYK2 inhibitors on sensitization of bortezomib-resistant MM cells in three *in vivo* models. In the first model, MM1s-Luc-GFP cells were injected intravenously into SCID mice and tumors were allowed to grow for 4 weeks; animals were then treated with a low-dose of bortezomib (0.5 mg/kg TIWx2) to induce resistance to bortezomib. The mice were divided into 6 groups and treated with (1) vehicle, (2) VS-4718, (3) VS-6063, (4) bortezomib, (5) combination of VS-4718 and bortezomib, and (6) combination of VS-6063 and bortezomib. Mouse survival was followed for four weeks. In the second model, H929 cells were injected subcutaneously (SC) into SCID mice; when tumors reached a mean of ~128 mm^3 mice were randomized into five groups and treated with (1) vehicle, (2) VS-4718, (3) bortezomib (1 mg/kg), (4) combination of VS-4718 and bortezomib (concurrently), and (5) combination of VS-4718 and bortezomib (sequentially to simulate minimum-residual disease). In the third model, H929 cells were injected SC into SCID mice; when tumors reached a mean of ~123 mm^3 mice were divided into 3 groups and treated with (1) vehicle, (2) bortezomib (0.9 mg/kg) and (3) combination of VS-6063 and bortezomib (sequentially to simulate minimum-residual disease). In all *in vivo* studies VS-4718 and VS-6063 were administered by oral gavage BID at 50 mg/kg.

Results

Figure 1: Dose-dependence effect of FAK/PYK2 inhibitors in normoxia and hypoxia

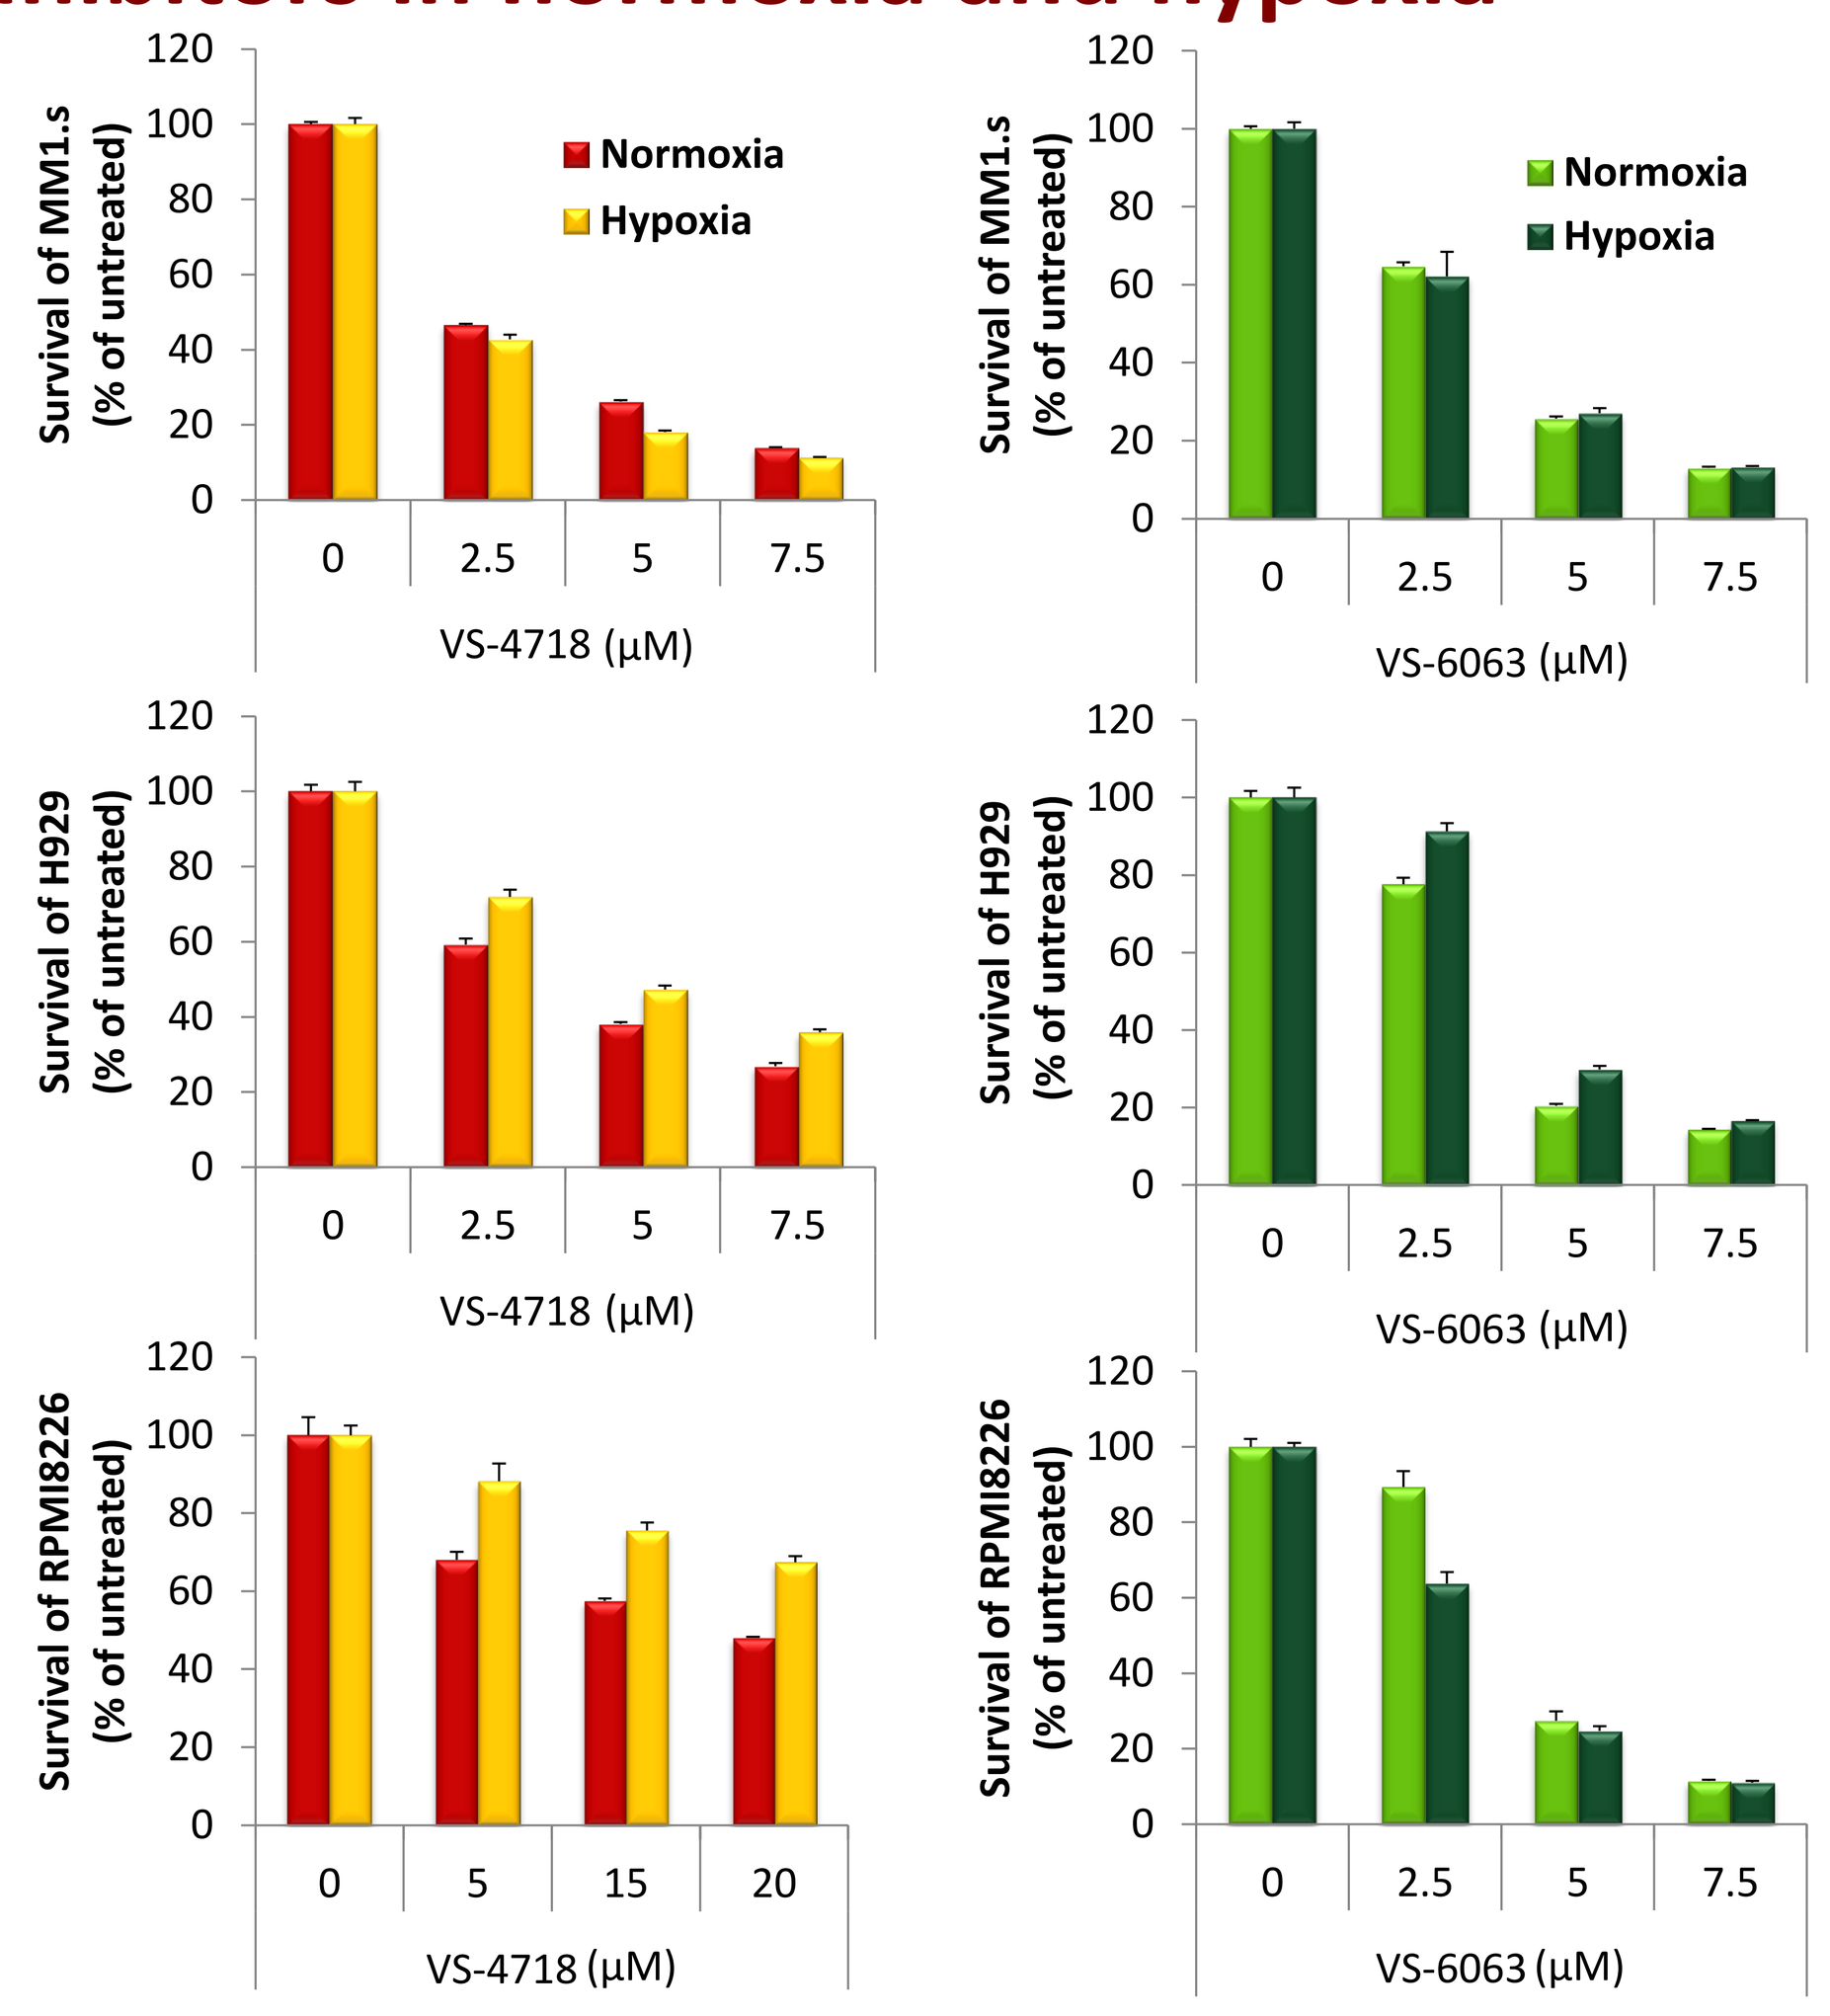
Normoxia
Hypoxia
Survival of MM1.s (% of untreated)
Survival of H929 (% of untreated)
Survival of RPMI8226 (% of untreated)
VS-4718 (µM)
VS-6063 (µM)

Figure 2: FAK/PYK2 inhibitors in combination with bortezomib reversed hypoxia-induced resistance in MM cells

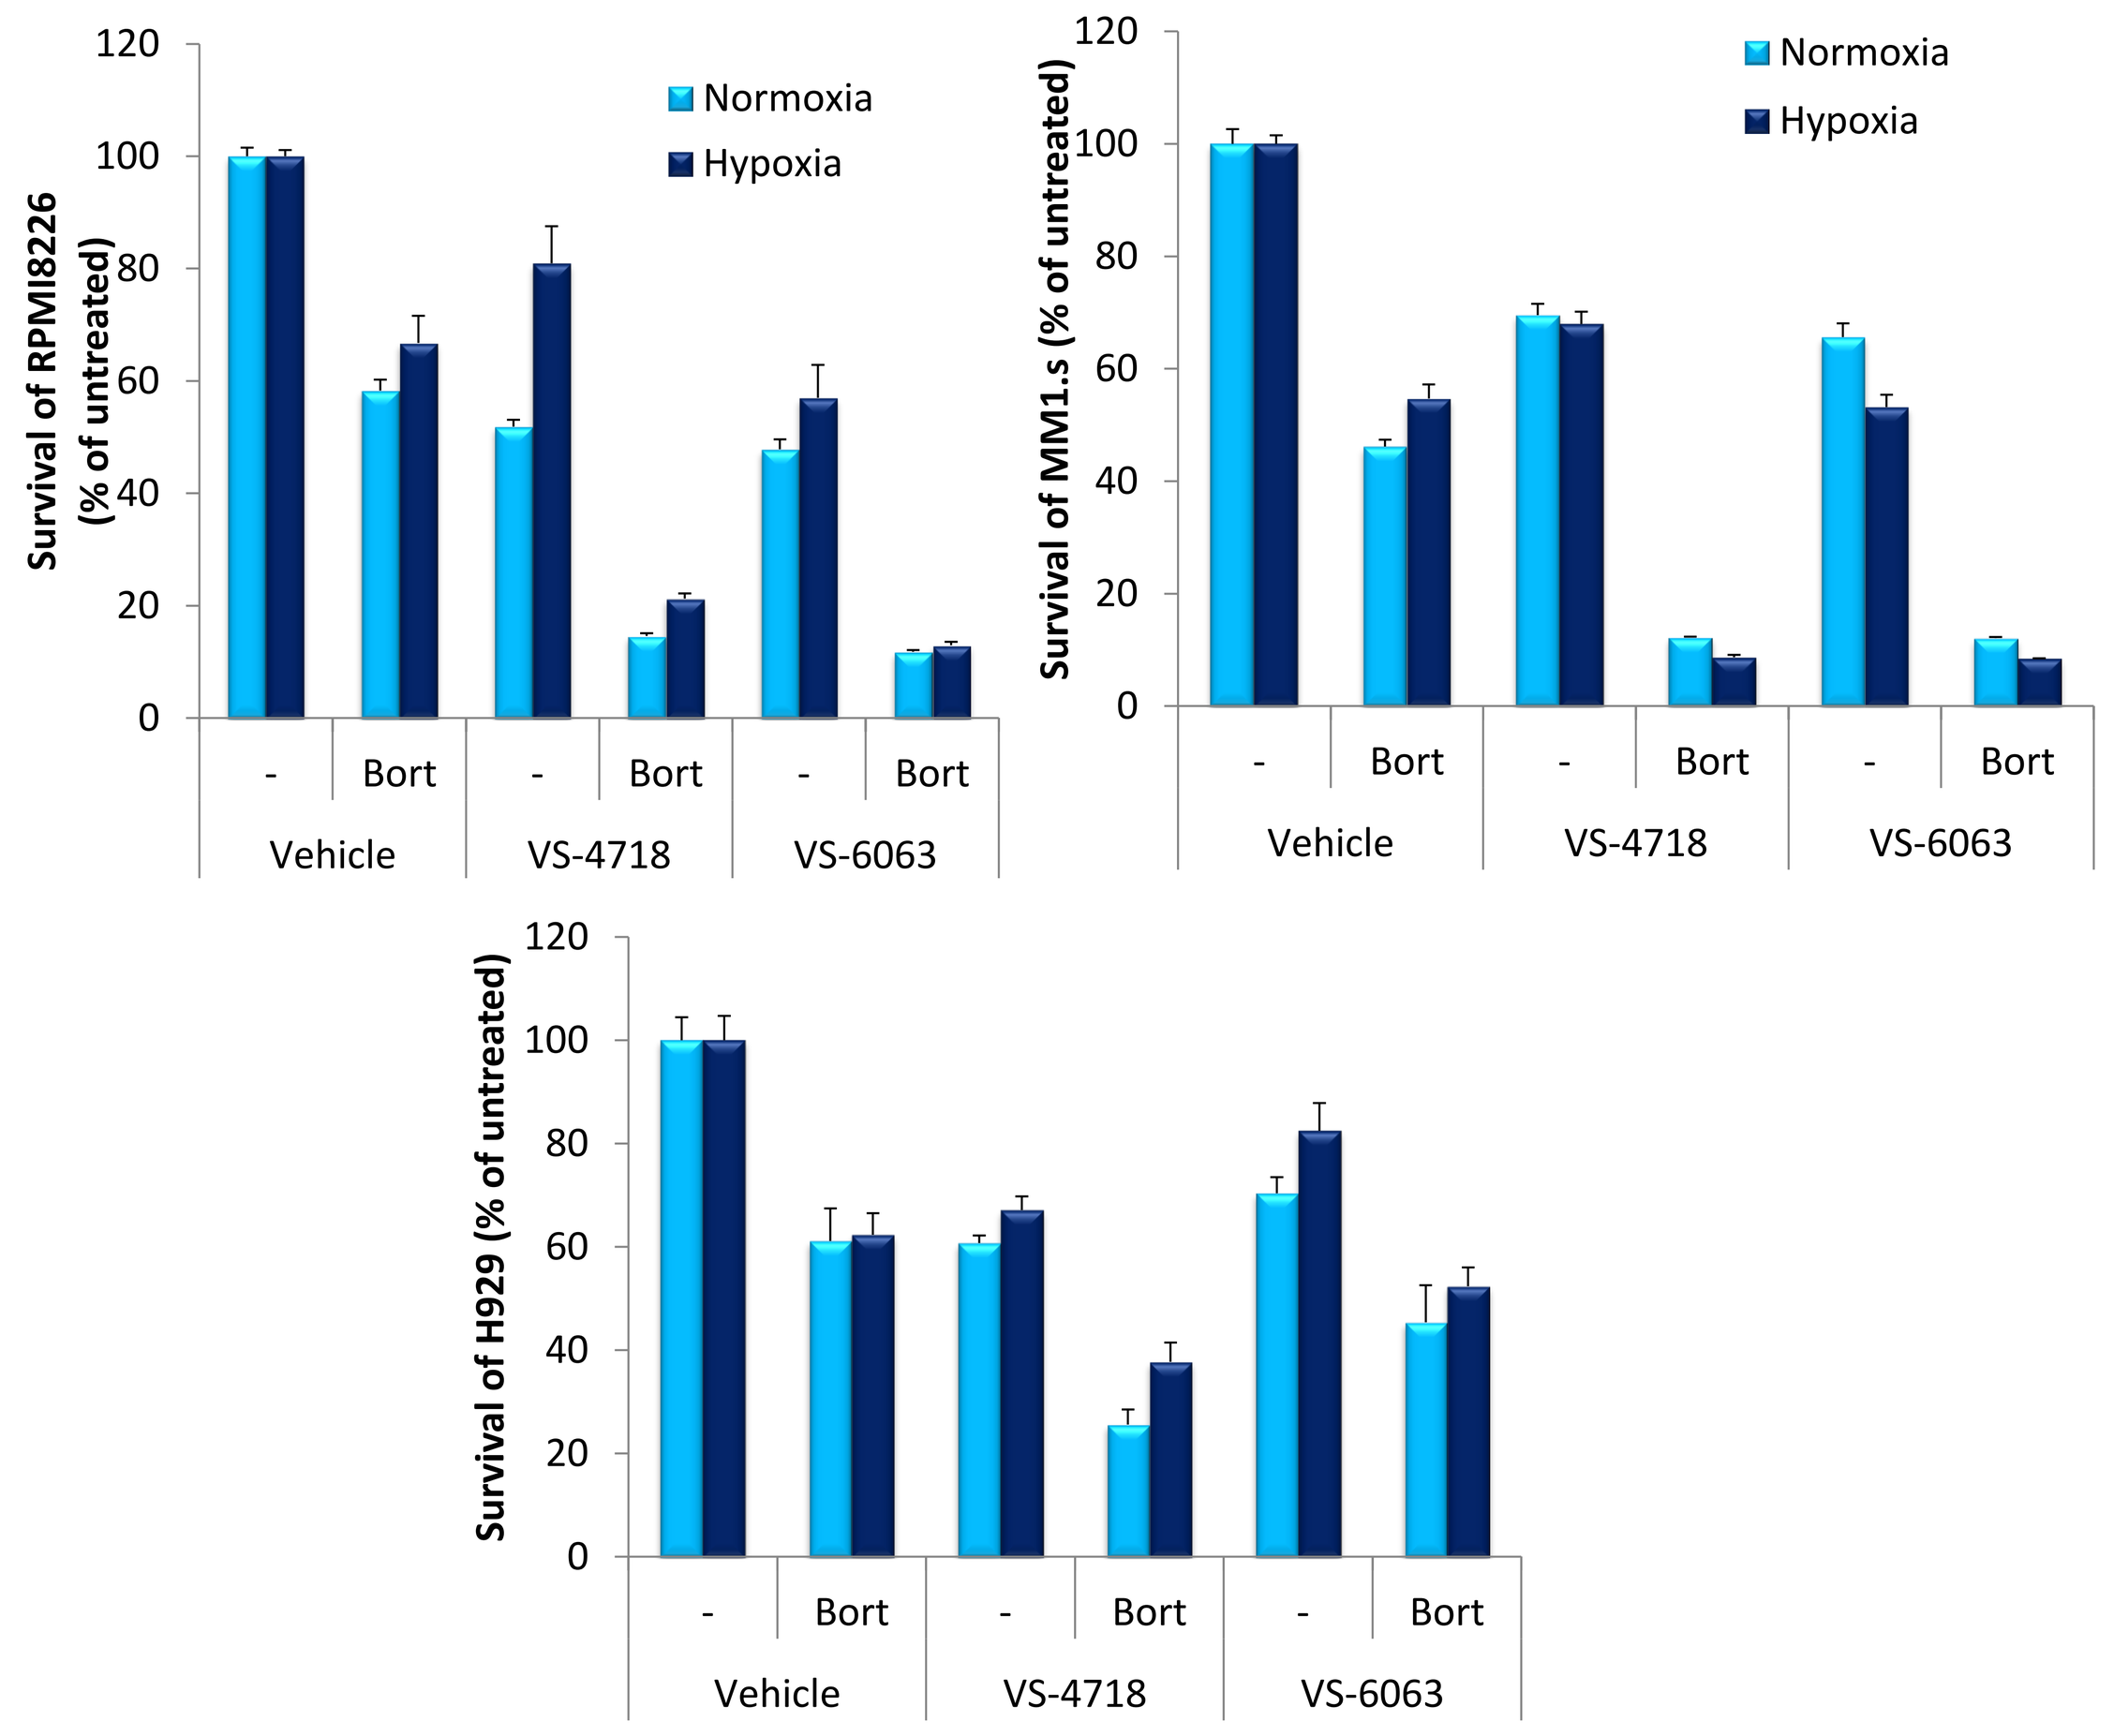
Normoxia
Hypoxia
Survival of RPMI8226 (% of untreated)
Survival of MM1.s (% of untreated)
Survival of H929 (% of untreated)
Vehicle
VS-4718
VS-6063
Bort

Figure 3: FAK/PYK2 inhibitors in combination with bortezomib induced MM cell apoptosis

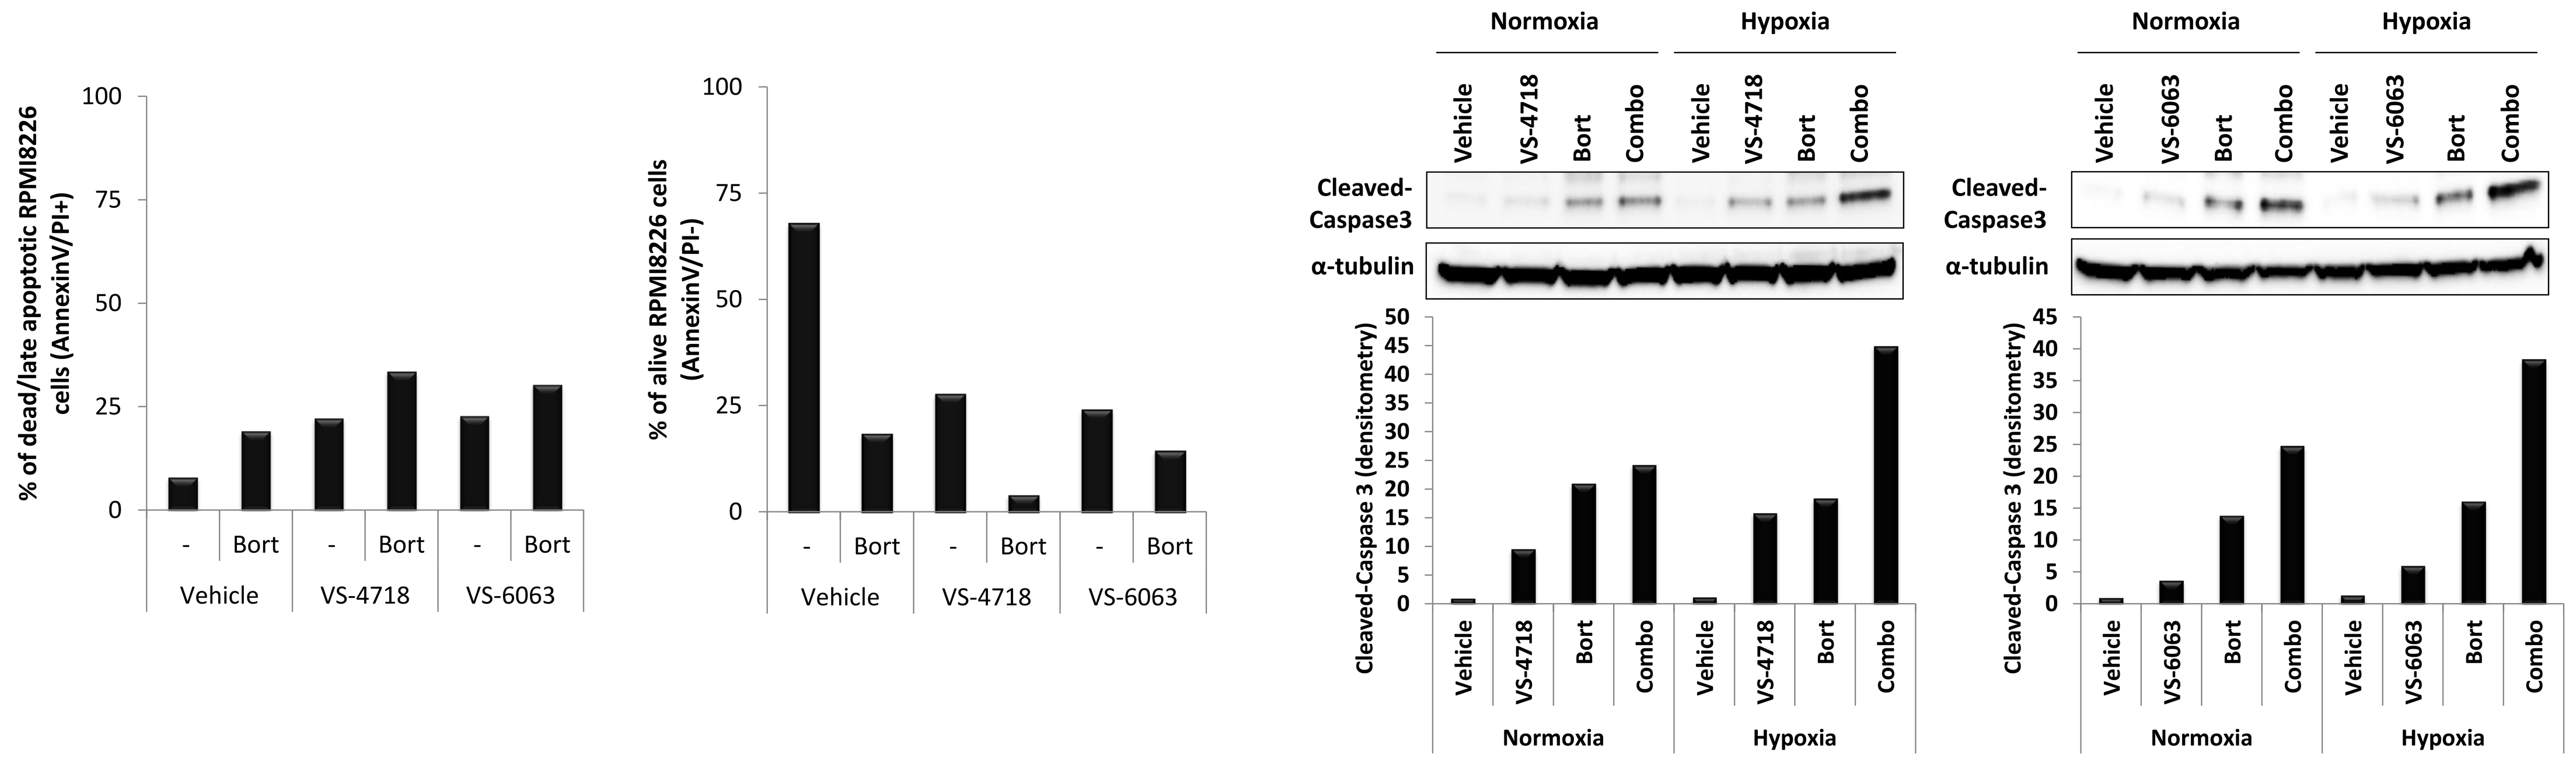
% of dead/late apoptotic RPMI8226 cells (AnnexinV/PI+)
% of alive RPMI8226 cells (AnnexinV/PI-)
Normoxia
Hypoxia
Vehicle
VS-4718
VS-6063
Bort
Combo
Cleaved-Caspase3
α-tubulin
Cleaved-Caspase 3 (densitometry)

Figure 4: FAK/PYK2 inhibitors resensitized MM cells *in vivo* when combined with bortezomib, and improved overall survival in mice (*in vivo* model 1)

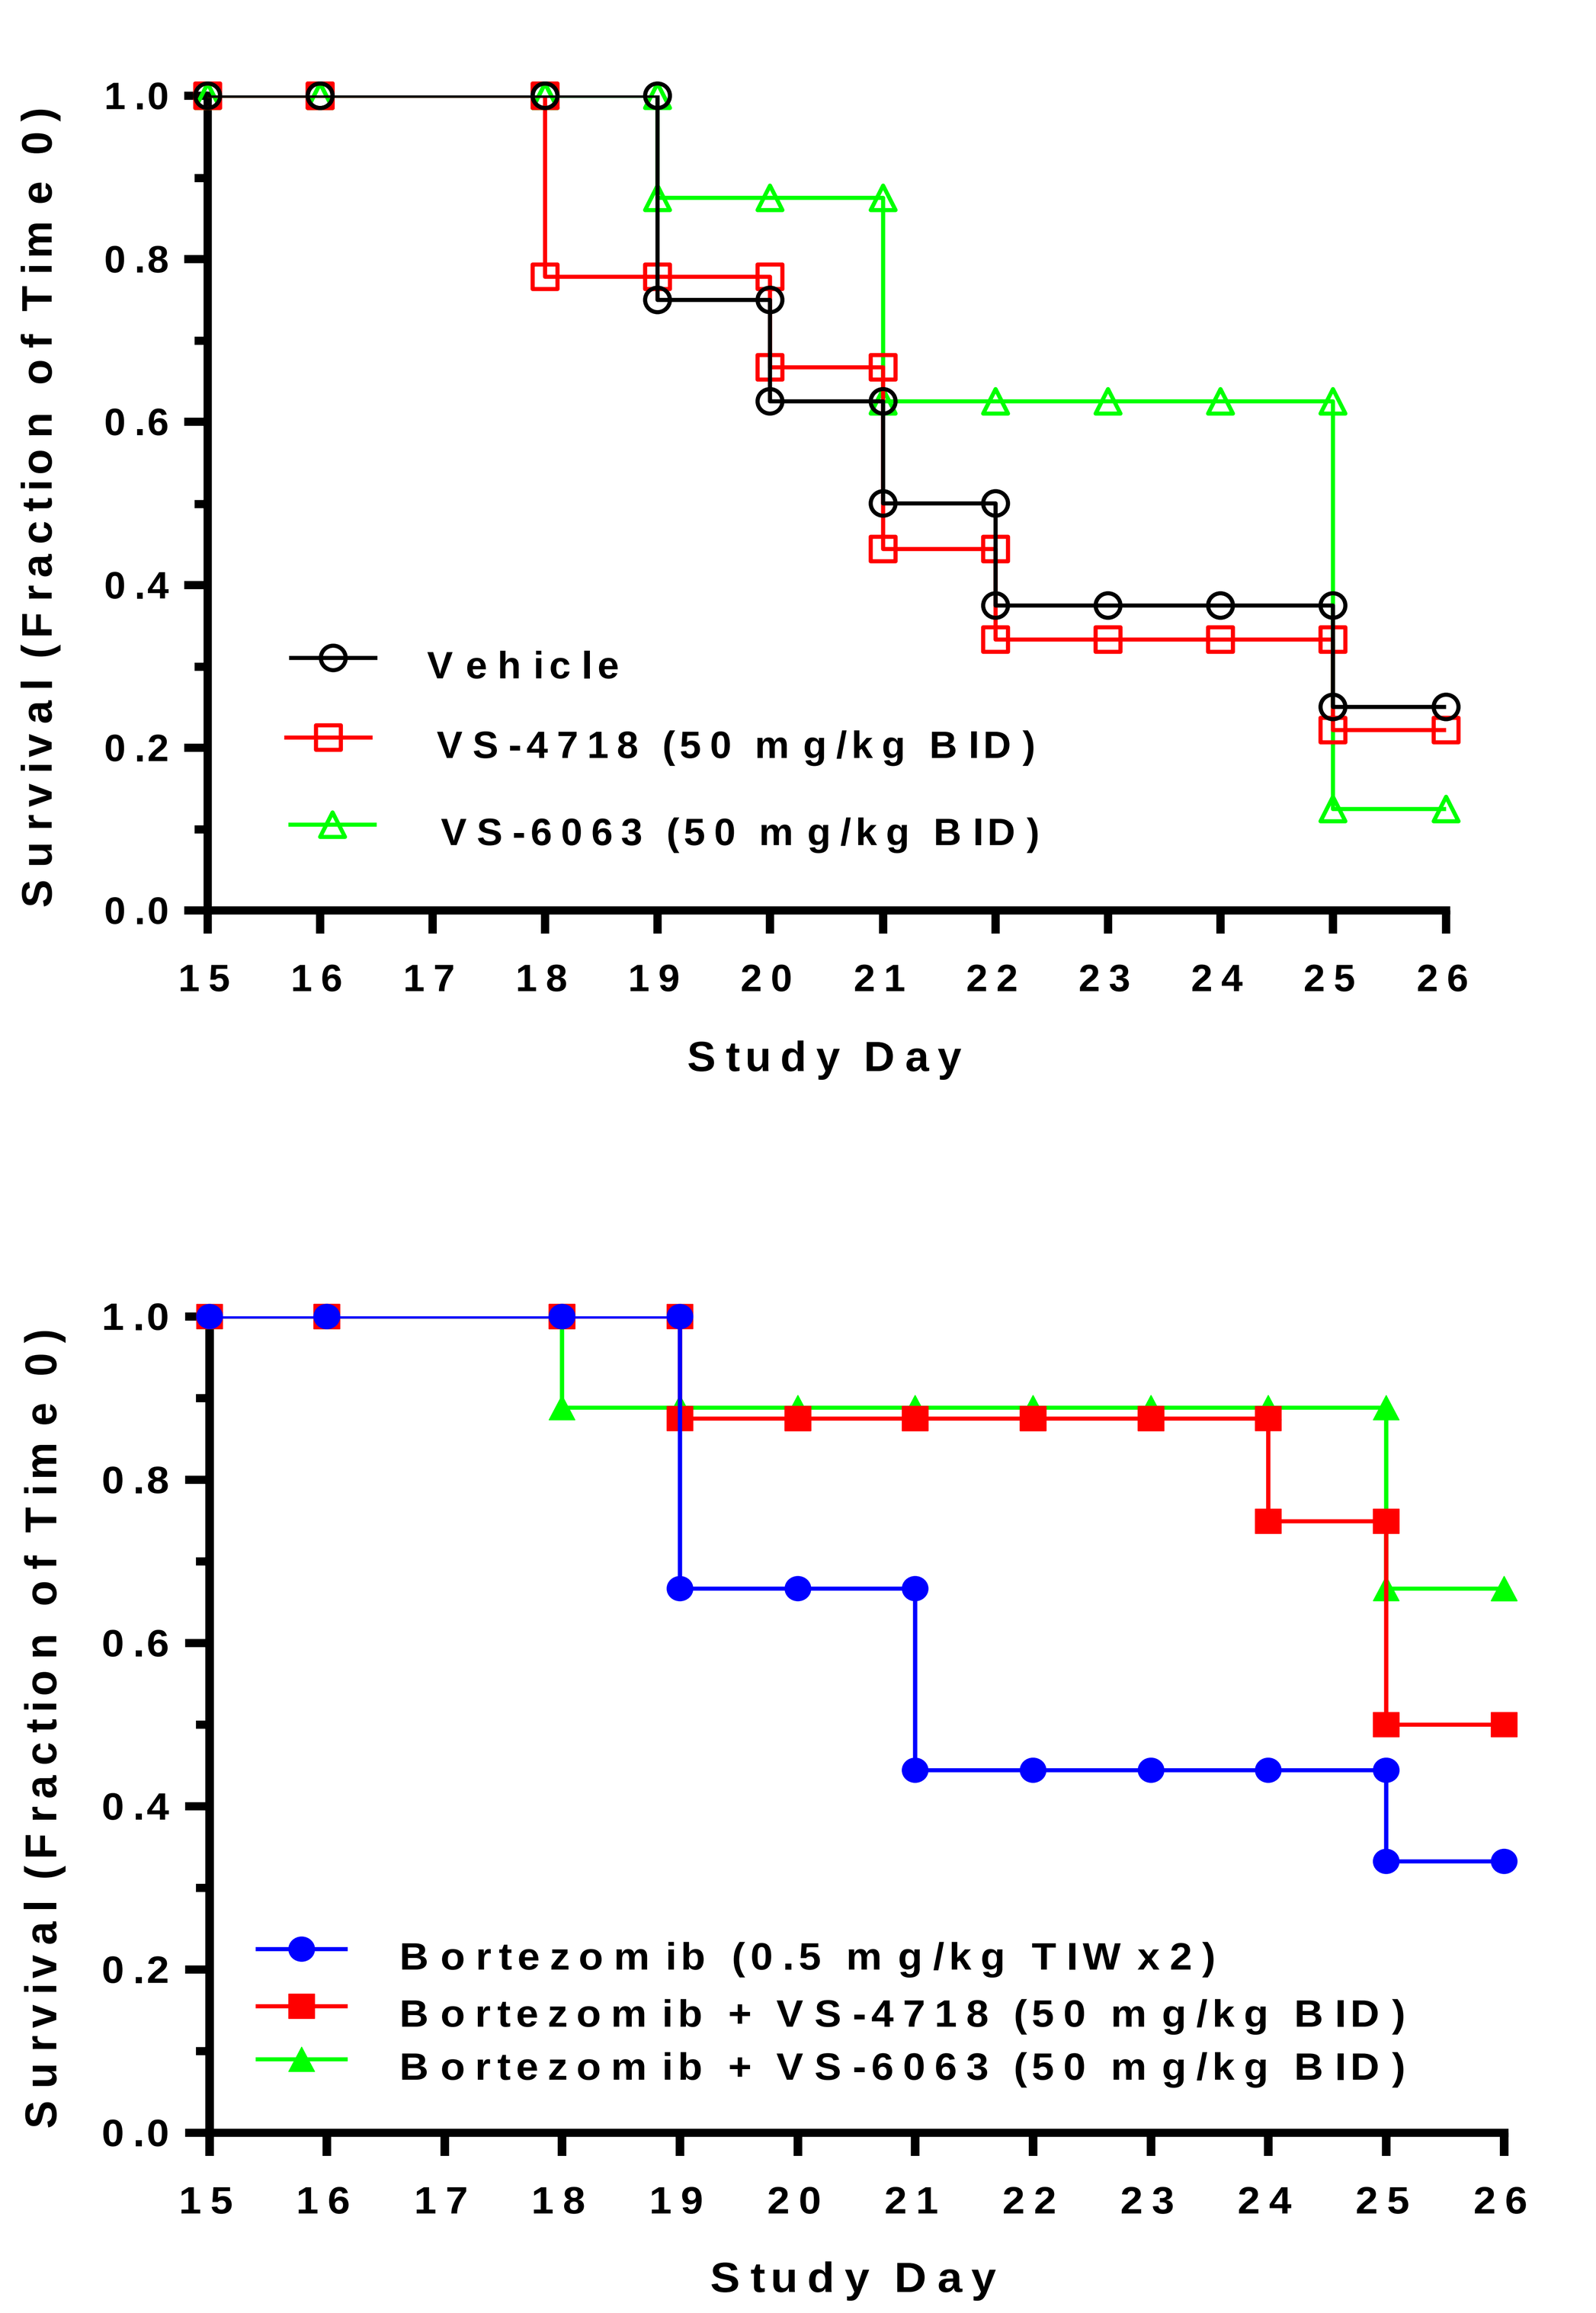
Survival (Fraction of Time 0)
Study Day
Vehicle
VS-4718 (50 mg/kg BID)
VS-6063 (50 mg/kg BID)
Bortezomib (0.5 mg/kg TIWx2)
Bortezomib + VS-4718 (50 mg/kg BID)
Bortezomib + VS-6063 (50 mg/kg BID)

Figure 5: FAK/PYK2 inhibitor (VS-4718) prevented MM relapse (*in vivo* model 2)

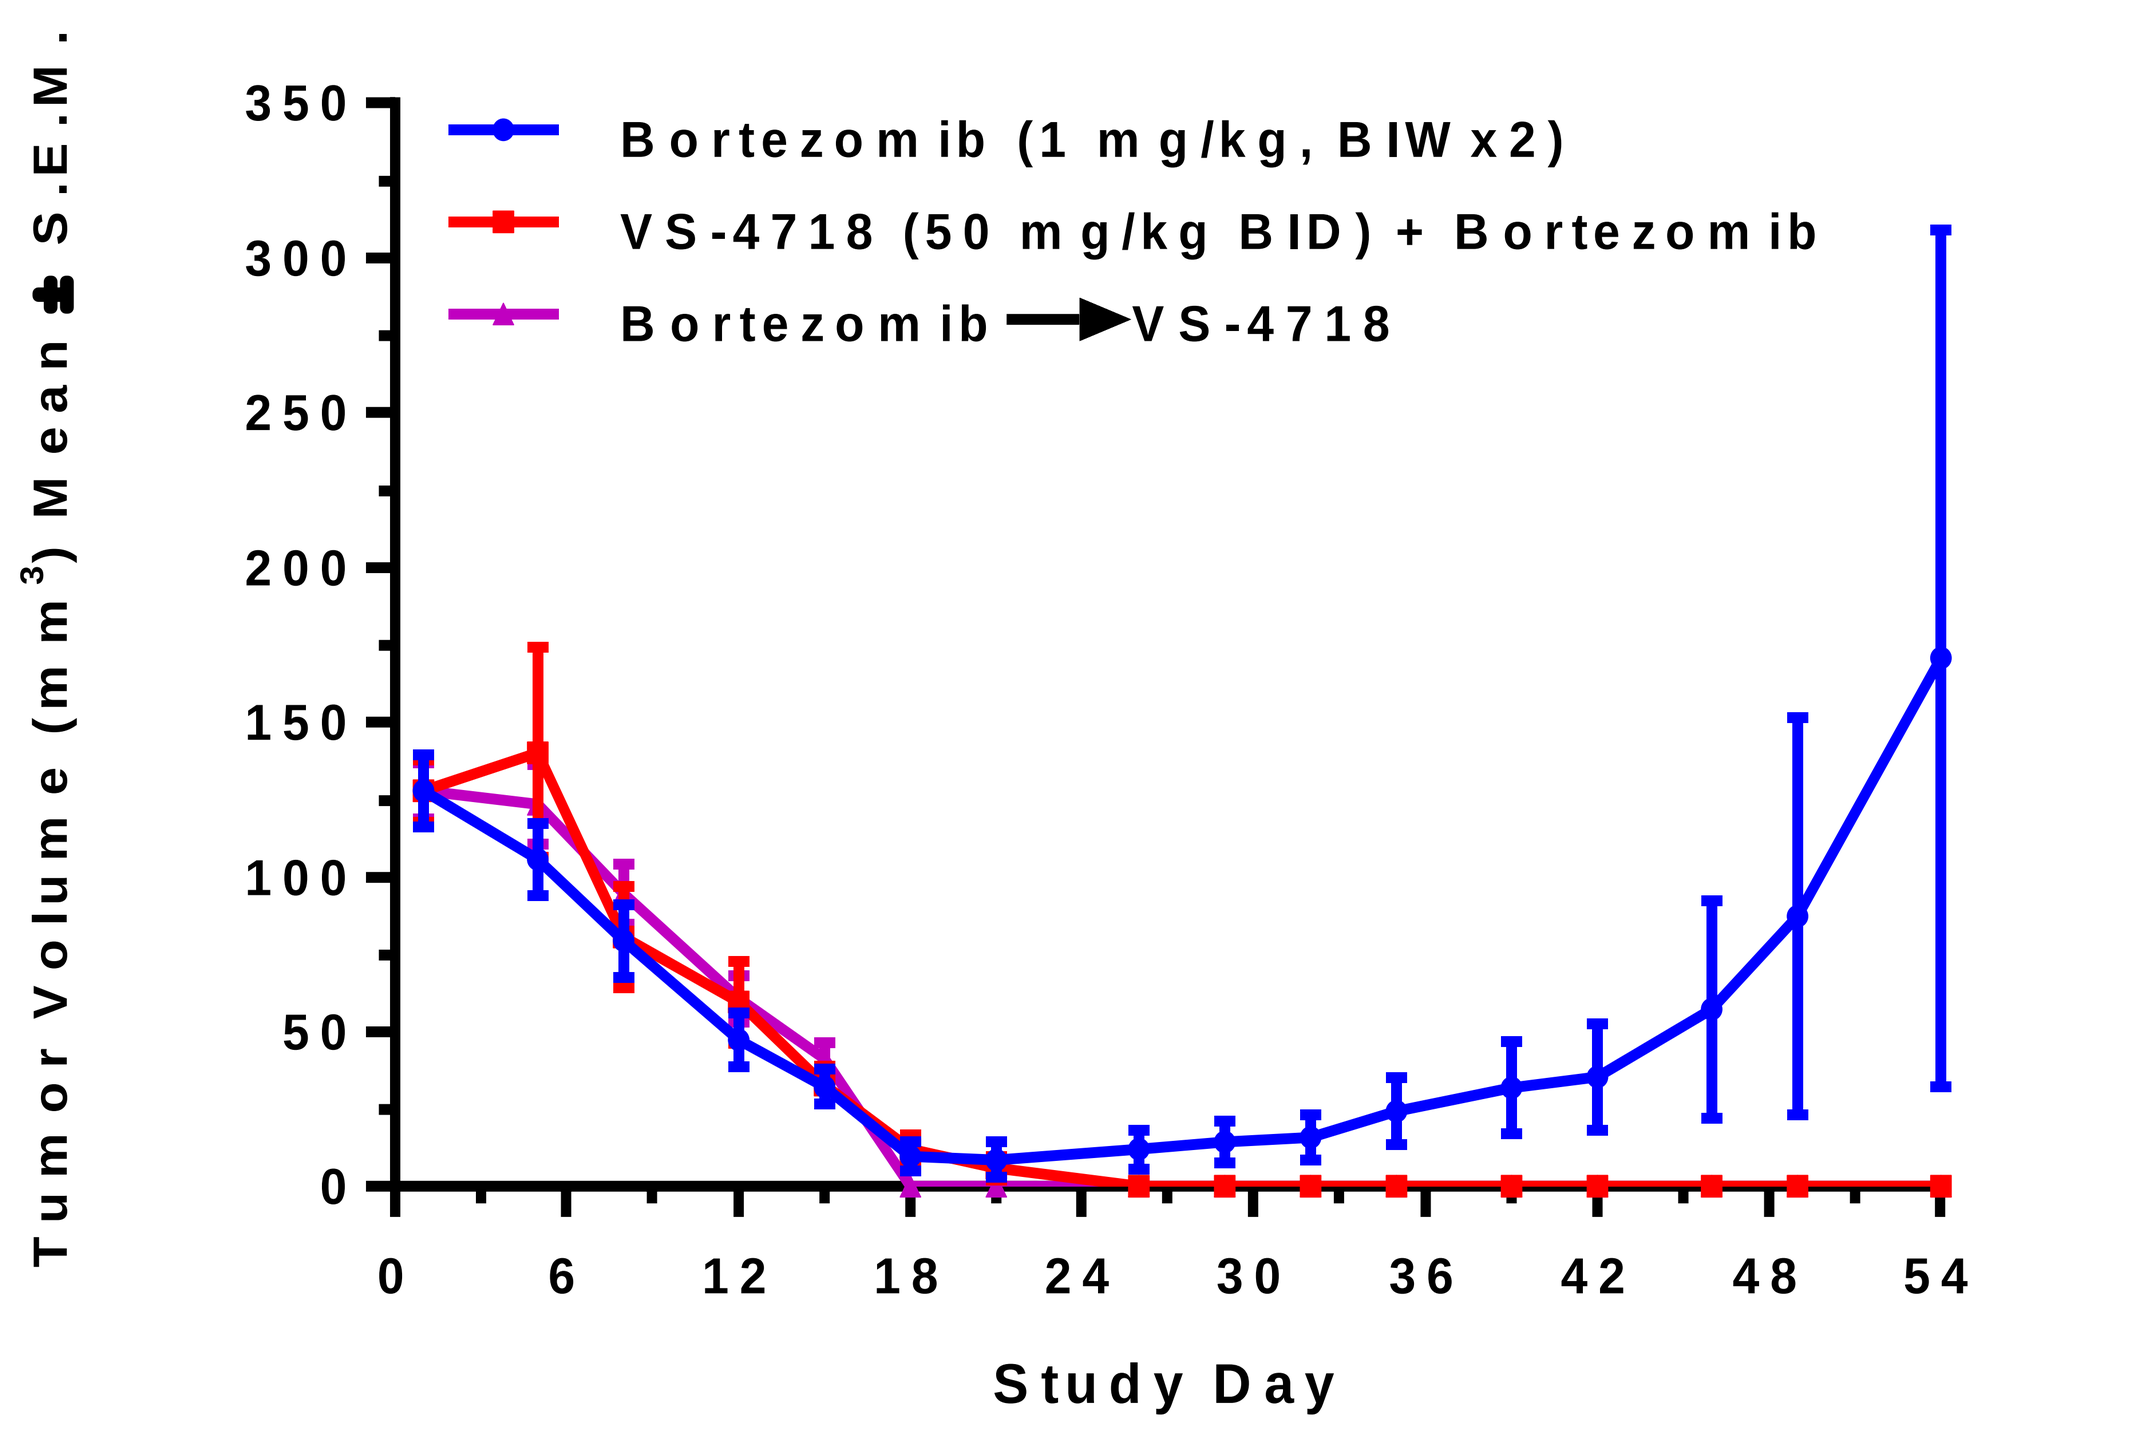
Tumor Volume (mm3) Mean ± S.E.M.
Study Day
Bortezomib (1 mg/kg, BIWx2)
VS-4718 (50 mg/kg BID) + Bortezomib
Bortezomib → VS-4718

Figure 6: FAK/PYK2 inhibitor (VS-6063) delayed MM relapse (*in vivo* model 3)

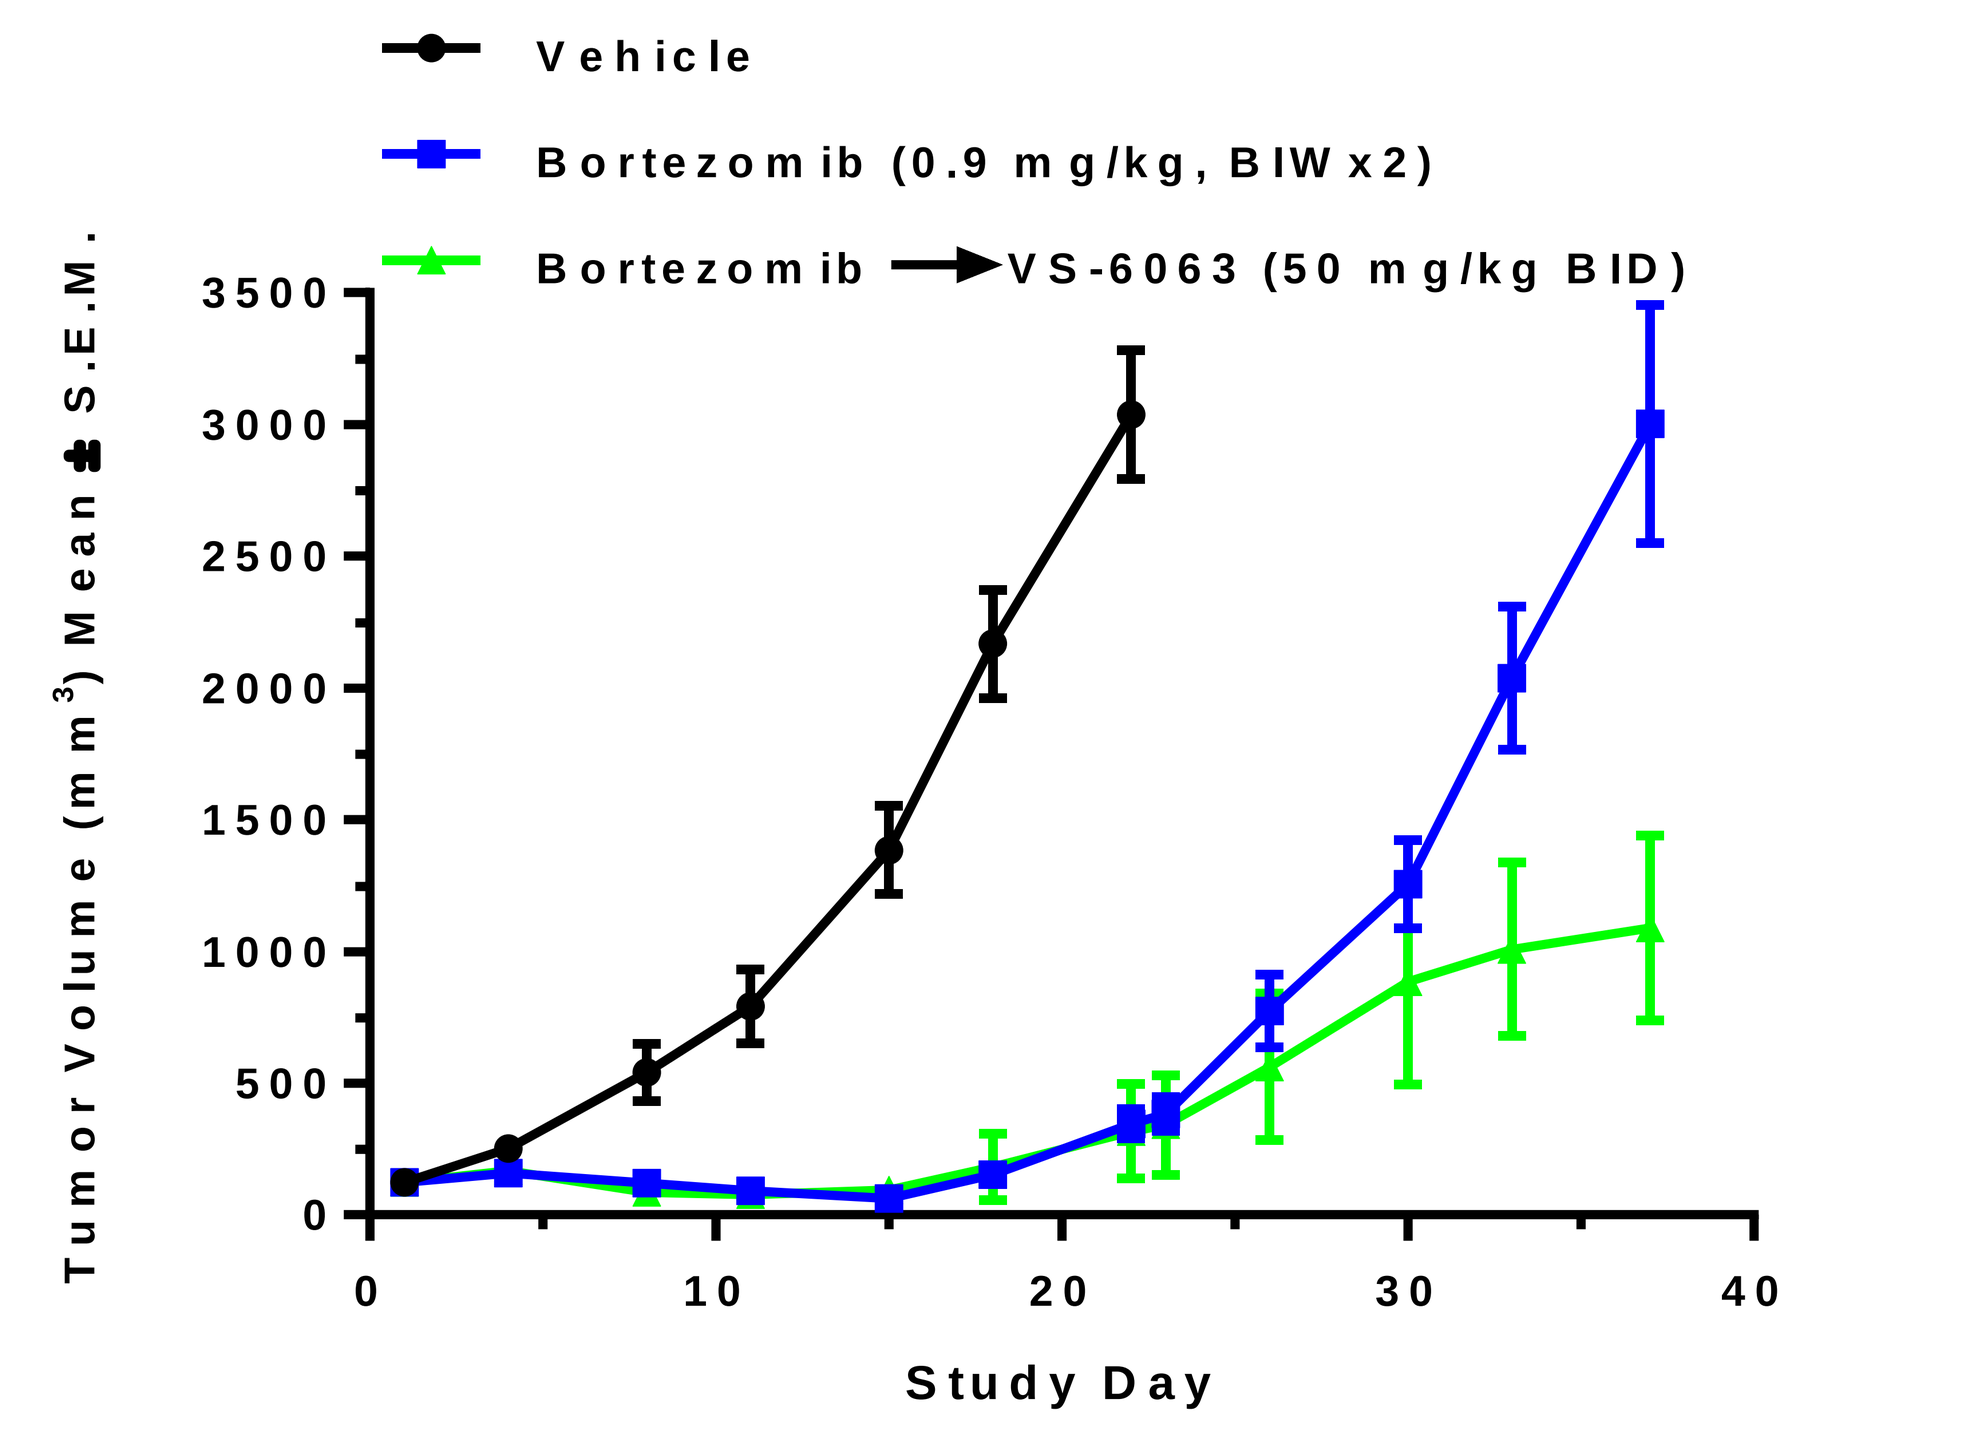
Vehicle
Bortezomib (0.9 mg/kg, BIWx2)
Bortezomib → VS-6063 (50 mg/kg BID)
Tumor Volume (mm3) Mean ± S.E.M.
Study Day

Conclusions

We report that FAK/PYK2 inhibitors, VS-4718 and VS-6063, decreased proliferation and increased apoptosis of MM cells as single agents. Hypoxia-induced resistance to proteasome inhibitors and the PYK2 inhibitors resensitized MM cells to therapy *in vitro* and *in vivo*. Moreover, FAK/PYK2 inhibitor VS-4718 was able to prevent relapse in an *in vivo* MM model simulating MRD. These data provide a basis for future clinical trials to sensitize relapsed/refractory MM patients to therapy by FAK/PYK2 inhibitors and their use to reduce relapse post front line treatment in an MRD setting. VS-6063 and VS-4718 are being evaluated in ongoing clinical trials for multiple types of cancer.

Contact Info

Corresponding author: Kareem Azab, B.Pharm, Ph.D.
Department of Radiation Oncology, Cancer Biology Division, Washington University in St Louis
e-mail: aazab@radonc.wustl.edu website: http://radonc.wustl.edu/research/cancerbio/azab/

COI: A.K. Azab reports receiving commercial research grant from Selexys, Verastem, Karyopharm and Cellworks.
No potential conflicts of interest were disclosed by other authors.