FAK/PYK2 INHIBITION ENHANCES IMMUNE CHECKPOINT INHIBITOR EFFICACY

Yan Wang[1], Jennifer E. Ring[1], Kam Sprott[1], David DeNardo[2], David T. Weaver[1] and Jonathan A. Pachter[1]

[1]Verastem Inc., Boston, MA; [2]Washington University, St. Louis, MO

Abstract # 568

ABSTRACT

Although durable responses to single agent immune checkpoint inhibitors have been reported, additional approaches are needed to extend this therapeutic benefit to a greater proportion of patients. Combination of immunotherapy agents with tumor microenvironment modulators has the potential to overcome barriers that tumor cells develop to evade the immune system, and provide benefit to a greater proportion of patients. Focal Adhesion Kinase (FAK) and the family member PYK2 are potentially valuable targets due to their roles in regulating key cellular populations in the tumor microenvironment. In addition to targeting cancer stem cells, the FAK/PYK2 dual inhibitors, VS-6063 and VS-4718, have been shown to inhibit monocyte-derived macrophages, reduce tumor-associated macrophages in xenograft models, and promote a CD8+ T cell-mediated anti-tumor response in squamous cell carcinoma models.

We now report the combination of VS-4718 with an anti-PD-1 mAb shows improved efficacy over anti-PD-1 mAb alone and extends survival of MC38 syngeneic tumor bearing animals. Analysis of MC38 tumors at day 12 of treatment revealed a significant increase in the CD8+ T cell/Treg ratio in tumors in the VS-4718 + anti-PD-1 combination group, providing a mechanistic understanding for the enhanced efficacy of this combination.

To explore additional combination options, we tested the combination of VS-4718 with anti-4-1BB in the MC38 model. Consistent with what was observed with the anti-PD-1 combination, VS-4718 also enhances the efficacy of an anti-4-1BB mAb.

To further delineate the effect of FAK inhibition, an *in vitro* T cell proliferation assay was conducted. VS-6063 and VS-4718 dose-dependently stimulated proliferation of CD8+ cytotoxic T cells isolated from healthy donors. This is in distinct contrast to other protein kinase inhibitors, such as the SRC inhibitor dasatinib which potently impaired the proliferation of CD8+ cytotoxic T cells. In addition, both VS-4718 and VS-6063 decreased CD8+ T cell exhaustion markers, and increased T cell-mediated tumor cell killing *in vitro*.

These data provide a rationale for clinical trials in cancer patients to test whether a FAK/PYK2 inhibitor in combination with an immune checkpoint inhibitor could increase the breadth of responsive tumor types, increase the number of responders, and confer more durable anti-tumor response.

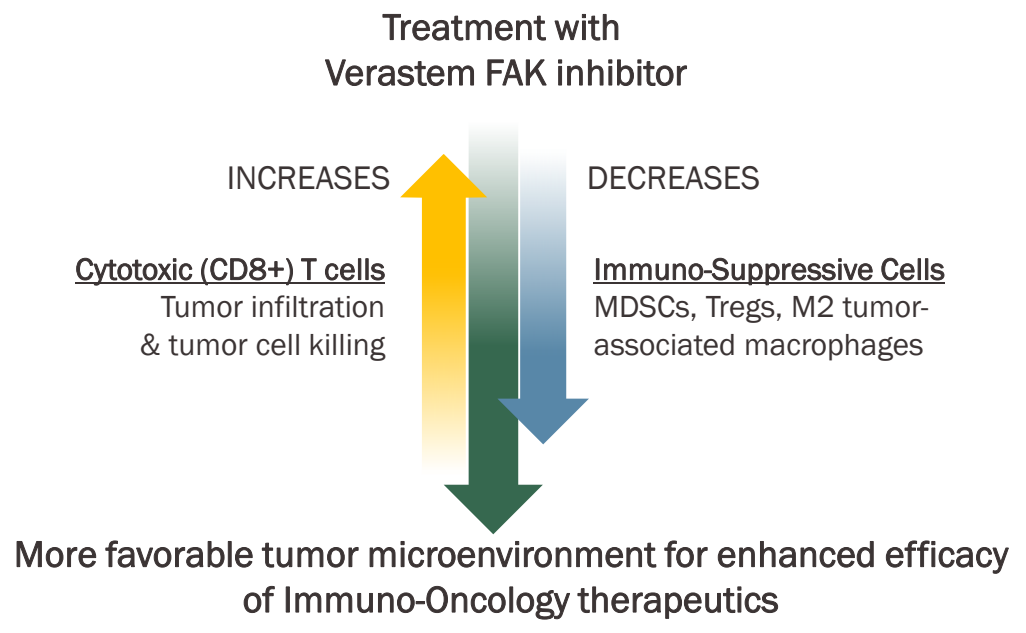
Treatment with
Verastem FAK inhibitor
INCREASES
DECREASES
Cytotoxic (CD8+) T cells
Tumor infiltration
& tumor cell killing
Immuno-Suppressive Cells
MDSCs, Tregs, M2 tumor-associated macrophages
More favorable tumor microenvironment for enhanced efficacy
of Immuno-Oncology therapeutics

FIGURE 1: Effects of FAK/PYK2 inhibitor on tumor microenvironment

RESULTS

Combination of VS-4718 + anti-PD1 improves anti-tumor efficacy in syngeneic MC38 colorectal tumor-bearing mice

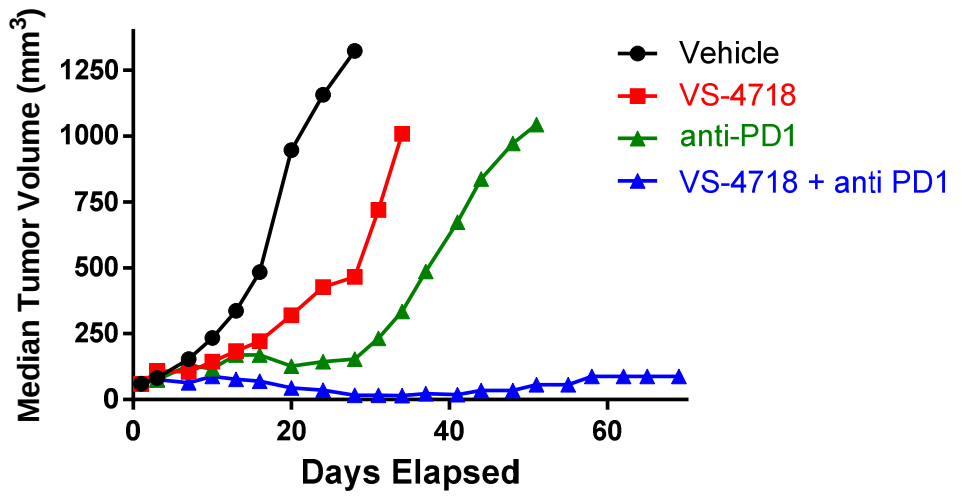
Median Tumor Volume (mm³)
Days Elapsed
Vehicle
VS-4718
anti-PD1
VS-4718 + anti PD1

FIGURE 2: Mice bearing syngeneic MC38 tumors were randomized once tumors reached 50-100mm³ and treated with either vehicle, VS-4718 (75 mg/kg, BID, po through end of experiment), anti-PD1 (clone RMP1-14, 10 mg/kg i.p. on days 1, 4, 8, 11) or VS-4718 + anti-PD1.

Decreased Tregs and increased CD8+ T cells with VS-4718 + anti-PD1 combination in MC38 Tumors

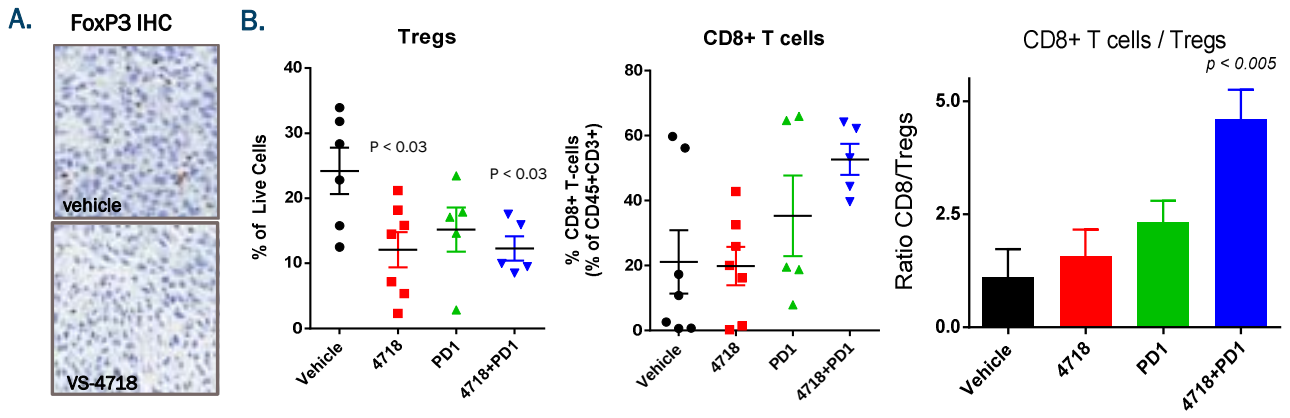
A.
FoxP3 IHC
vehicle
VS-4718
B.
Tregs
% of Live Cells
P < 0.03
P < 0.03
CD8+ T cells
% CD8+ T-cells (% of CD45+CD3+)
CD8+ T cells / Tregs
p < 0.005
Ratio CD8/Tregs
Vehicle
4718
PD1
4718+PD1

FIGURE 3: Syngeneic MC38 tumors were randomized once tumors reached 50-100mm³ and then treated with either vehicle, VS-4718 (50 mg/kg, BID), anti-PD1 (clone RMP1-14, 5 mg/kg on days 1, 4, 8, 11) or VS-4718 + anti-PD1 for 12 days at which point tumors were harvested and embedded in FFPE for staining FoxP3 by IHC [A] or processed live for flow cytometry [B].

Combination of VS-4718 + anti-4-1BB improves anti-tumor efficacy in syngeneic MC38 colorectal tumor-bearing mice

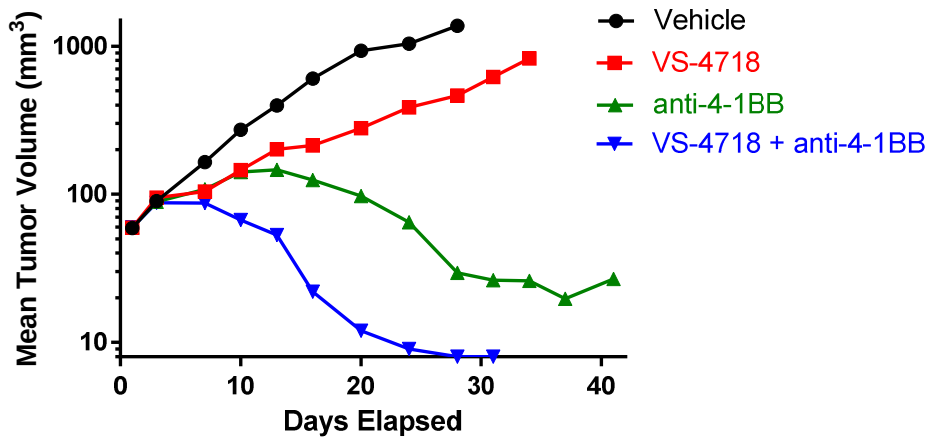
Mean Tumor Volume (mm³)
Days Elapsed
Vehicle
VS-4718
anti-4-1BB
VS-4718 + anti-4-1BB

FIGURE 4: Syngeneic MC38 tumors were randomized once tumors reached 50-100mm³ and then treated with either vehicle, VS-4718 (75 mg/kg, BID, po), anti-4-1BB (clone LOB12.3, 10 mg/kg i.p. on days 1, 4, 8, 11) or VS-4718 + anti-4-1BB.

VS-4718 enhances CD8+ T cell killing of tumor cells

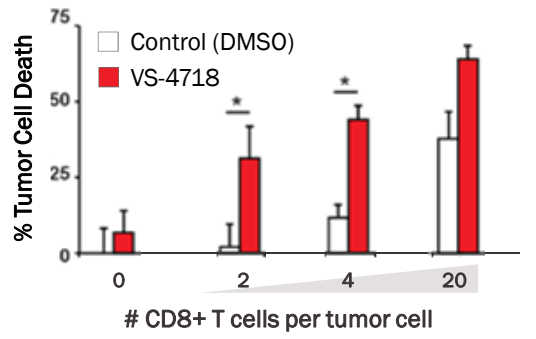
% Tumor Cell Death
Control (DMSO)
VS-4718
CD8+ T cells per tumor cell

FIGURE 5: Antigen-specific CD8+ T cells were co-cultured with cognate luciferized tumor cells at various E:T ratios in the presence of VS-4718 1uM on top of Matrigel. After 24 hours, cultured were analyzed for changes in luciferase as a measure of tumor cell viability.

VS-4718 and VS-6063 enhance primary CD8+ T cell proliferation

Cellular EC_{50} (nM) Avg ± SD

	FAK	PYK2
VS-4718	34 ± 27	43 ± 40
VS-6063	21 ± 4	46 ± 37

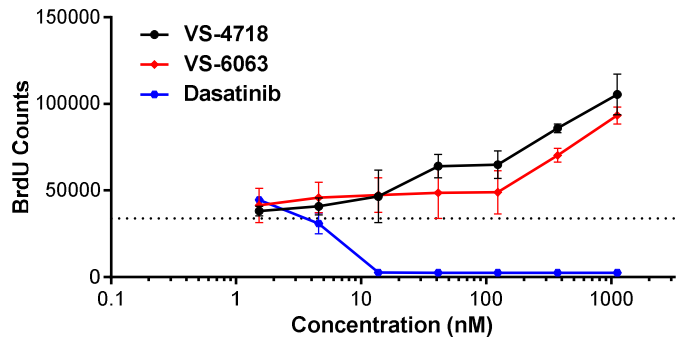
BrdU Counts
VS-4718
VS-6063
Dasatinib
Concentration (nM)

FIGURE 6: CD8+ T cells were isolated from fresh, healthy human PBMCs by negative immunomagnetic bead separation. Purified CD8+ T cells were plated on CD3-coated plates in the presence or absence of VS-4718 or VS-6063 for 72 hours. Assay wells were pulsed with BrdU for the last 3-4 hours of culture and subjected to a BrdU-incorporation assay for the determination of actively proliferating cells. Data are presented as fold-change vs. DMSO control wells.

VS-4718 and VS-6063 decrease CD8+ T cell exhaustion markers

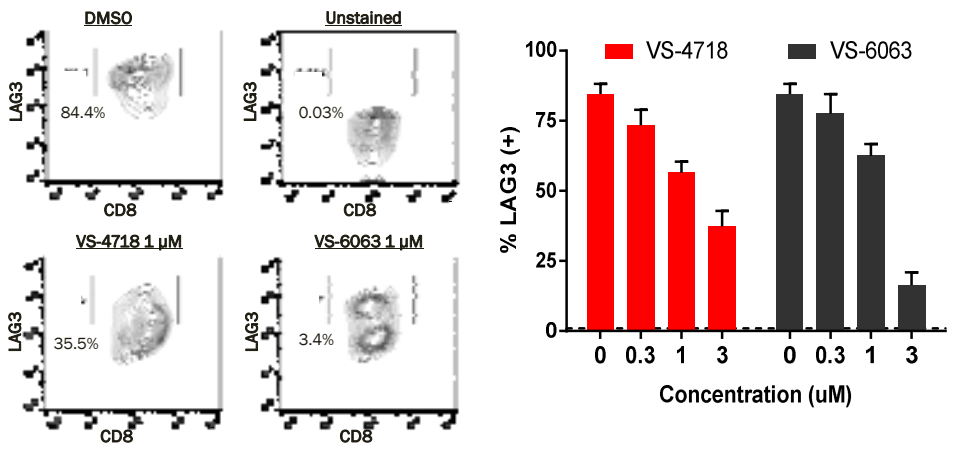
DMSO
84.4%
Unstained
0.03%
VS-4718 1 µM
35.5%
VS-6063 1 µM
3.4%
LAG3
CD8
% LAG3 (+)
VS-4718
VS-6063
Concentration (uM)

FIGURE 7: CD8+ T cells isolated from fresh, healthy human PBMCs by negative immunomagnetic bead separation were plated on anti-CD3 coated plates in the presence of VS-4718 or VS-6063 for 72 hours and then harvested and stained with anti-LAG3 or anti-PD-1 for flow cytometric analysis

SUMMARY

- FAK inhibitors change the tumor immune balance to potentiate efficacy of various immuno-oncology agents
 - FAK inhibitor combination substantially enhances anti-tumor efficacy of anti-PD-1 or anti-4-1BB vs. each immuno-oncology antibody alone
- FAK inhibitor + anti-PD-1 combination decreases Tregs and increases CD8+ T cells in MC38 tumors
- FAK inhibitors increase CD8+ T cell proliferation, decrease CD8+ T cell exhaustion markers, and increase T cell-mediated tumor cell killing *in vitro*
- These data provide rationale for clinical use of a FAK inhibitor in combination with immuno-oncology antibodies
 - Supports the ongoing VS-6063 combination trial with pembrolizumab (anti-PD-1) and gemcitabine in pancreatic cancer (NCT02546531) & the clinical testing of VS-6063 + avelumab (anti-PD-L1) in ovarian cancer

Verastem
novel drugs targeting cancer stem cells